Further Characterization of a Novel AAV Vector and Expanded Selection Criteria Platform for Muscle Gene Delivery

Jennifer Green, Jessica F Boehler, Tiffany Willacy, Prushti Bhavsar, J Patrick Gonzalez, Kristy J Brown, Carl Morris

Solid Biosciences, Charlestown, MA, USA

Introduction

- For Duchenne muscular dystrophy (DMD), gene therapy is under investigation to replace the absent dystrophin with a smaller, functional microdystrophin (µDys).
- Since muscle constitutes a large proportion of body mass, a high dose of vector administered systemically is necessary. Identification and development of more muscle-tropic vectors may improve safety and efficacy of DMD gene therapies.
- A lead novel AAV capsid of interest (AAV-SLB101) has increased muscle tropism and decreased liver transduction in disease and non-disease contexts in multiple animal models (*DMD[mdx]* mice, wild type mice and non-human primates).
- Based on our hypothesized mechanism of transduction of this lead candidate, additional novel AAV capsids have been designed and tested.

Novel Capsid in Mouse Models

DMD[mdx] *mice*

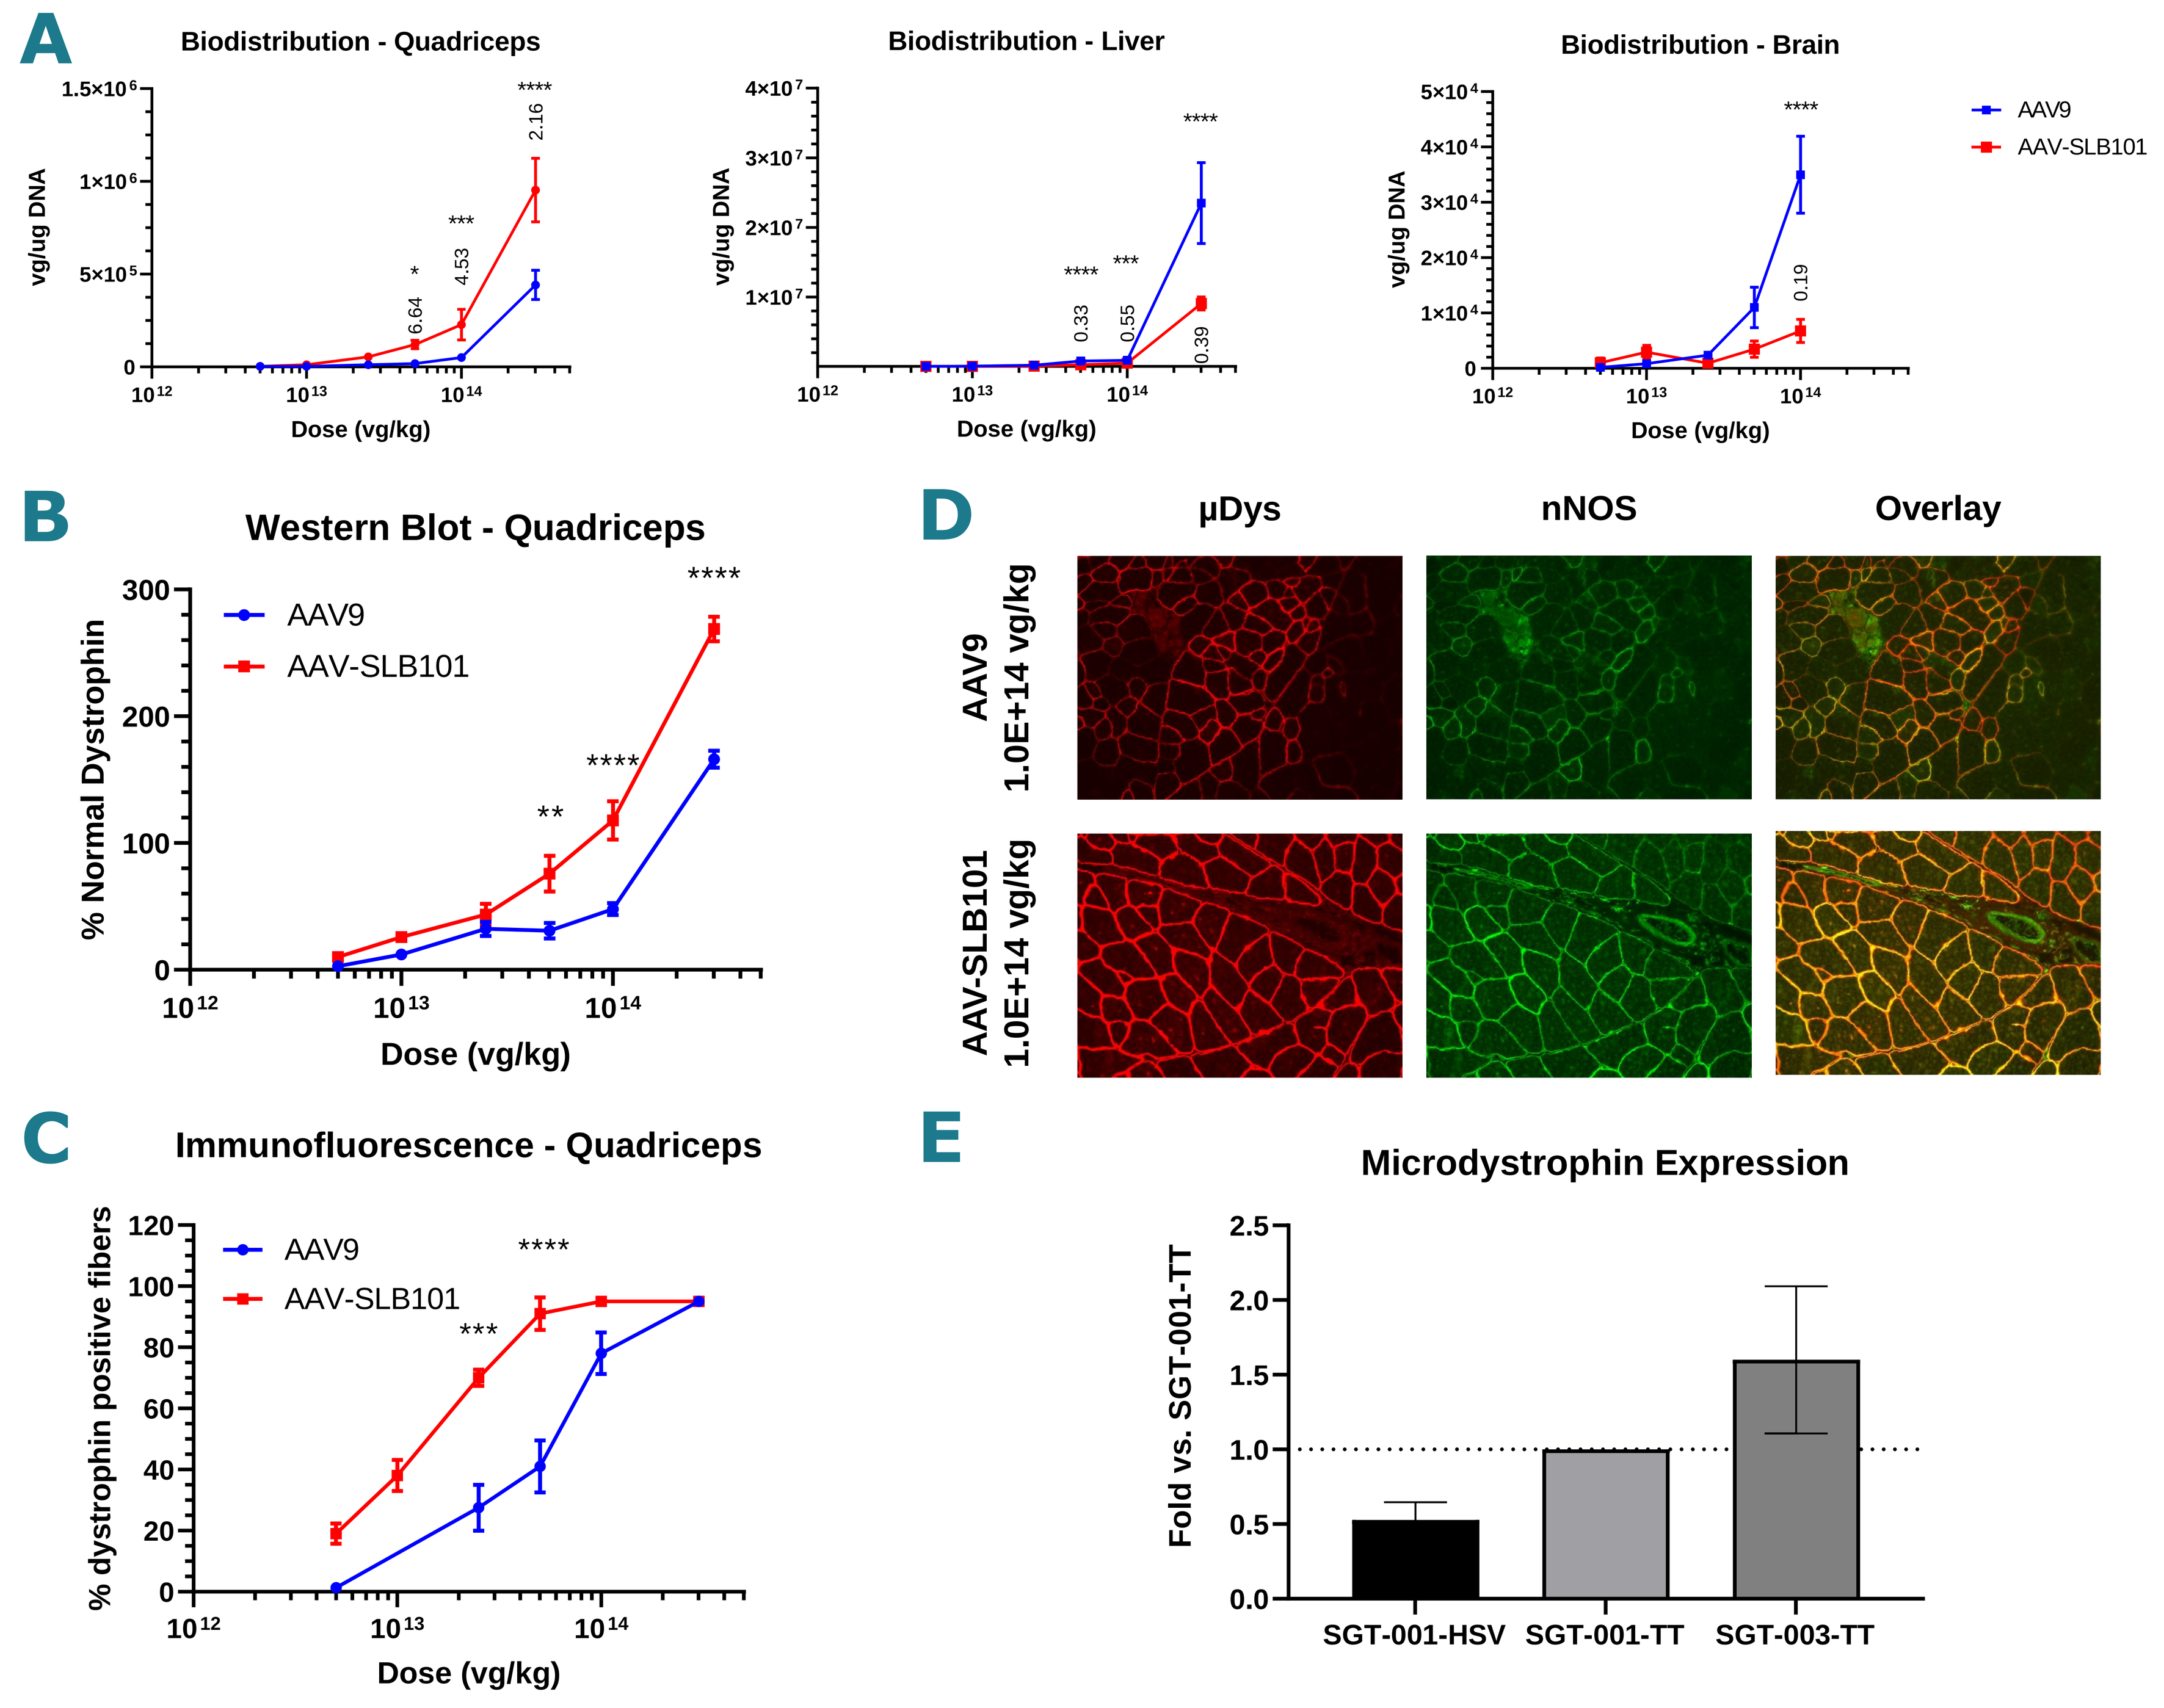
A
Biodistribution - Quadriceps
Biodistribution - Liver
Biodistribution - Brain
vg/ug DNA
Dose (vg/kg)
AAV9
AAV-SLB101
6.64
4.53
2.16
0.33
0.55
0.39
0.19
B
Western Blot - Quadriceps
% Normal Dystrophin
C
Immunofluorescence - Quadriceps
% dystrophin positive fibers
D
µDys
nNOS
Overlay
AAV9 1.0E+14 vg/kg
AAV-SLB101 1.0E+14 vg/kg
E
Microdystrophin Expression
Fold vs. SGT-001-TT
SGT-001-HSV
SGT-001-TT
SGT-003-TT

Figure 1. *DMD[mdx]* mice, 5-6 weeks old, were systemically injected with AAV9 and AAV-SLB101 at six doses ranging from 5E12 to 3E14 vg/kg. Mice were necropsied four weeks post-injection and tissues were harvested. Differentiated biodistribution and microdystrophin expression were observed in comparison to AAV9. Statistics in all panels were determined by ordinary two-way ANOVA.

A. AAV biodistribution by qPCR. Increased biodistribution of AAV-SLB101 over AAV9 to the quadriceps was observed at 5E13 vg/kg and above. Inversely, decreased biodistribution of AAV-SLB101 to the liver and brain at higher doses in comparison to AAV9 was observed. Fold change relative to AAV9 is indicated when difference was statistically significant (* $p < 0.05$, *** $p < 0.001$ and **** $p < 0.0001$).
B. Microdystrophin protein expression by western blot. AAV-SLB101 had significantly higher µDys expression compared to AAV9 at 5E13 vg/kg and above (** $p < 0.01$, **** $p < 0.0001$).
C. Microdystrophin protein expression by immunofluorescence. AAV-SLB101 had significantly higher µDys expression compared to AAV9 at 2.5E13 and 5E13 vg/kg (*** $p < 0.001$, **** $p < 0.0001$).
D. Representative immunofluorescence of microdystrophin (red), nNOS (green) and overlays from either 2.5E13 or 1E14 vg/kg of AAV9 or AAV-SLB101.
E. Relative microdystrophin protein expression by western blot across multiple in vivo evaluations of SGT-001 (rAAV9-CK8-µDys) manufactured by either HSV-based or transient transfection (TT)-based methodology and SGT-003 (rAAV-SLB101-CK8-µDys) manufactured by TT-based methodology. Additive improvements in expression were observed in both manufacturing and capsid for the SGT-003 drug candidate.

Wild type mice (C57BL/6)

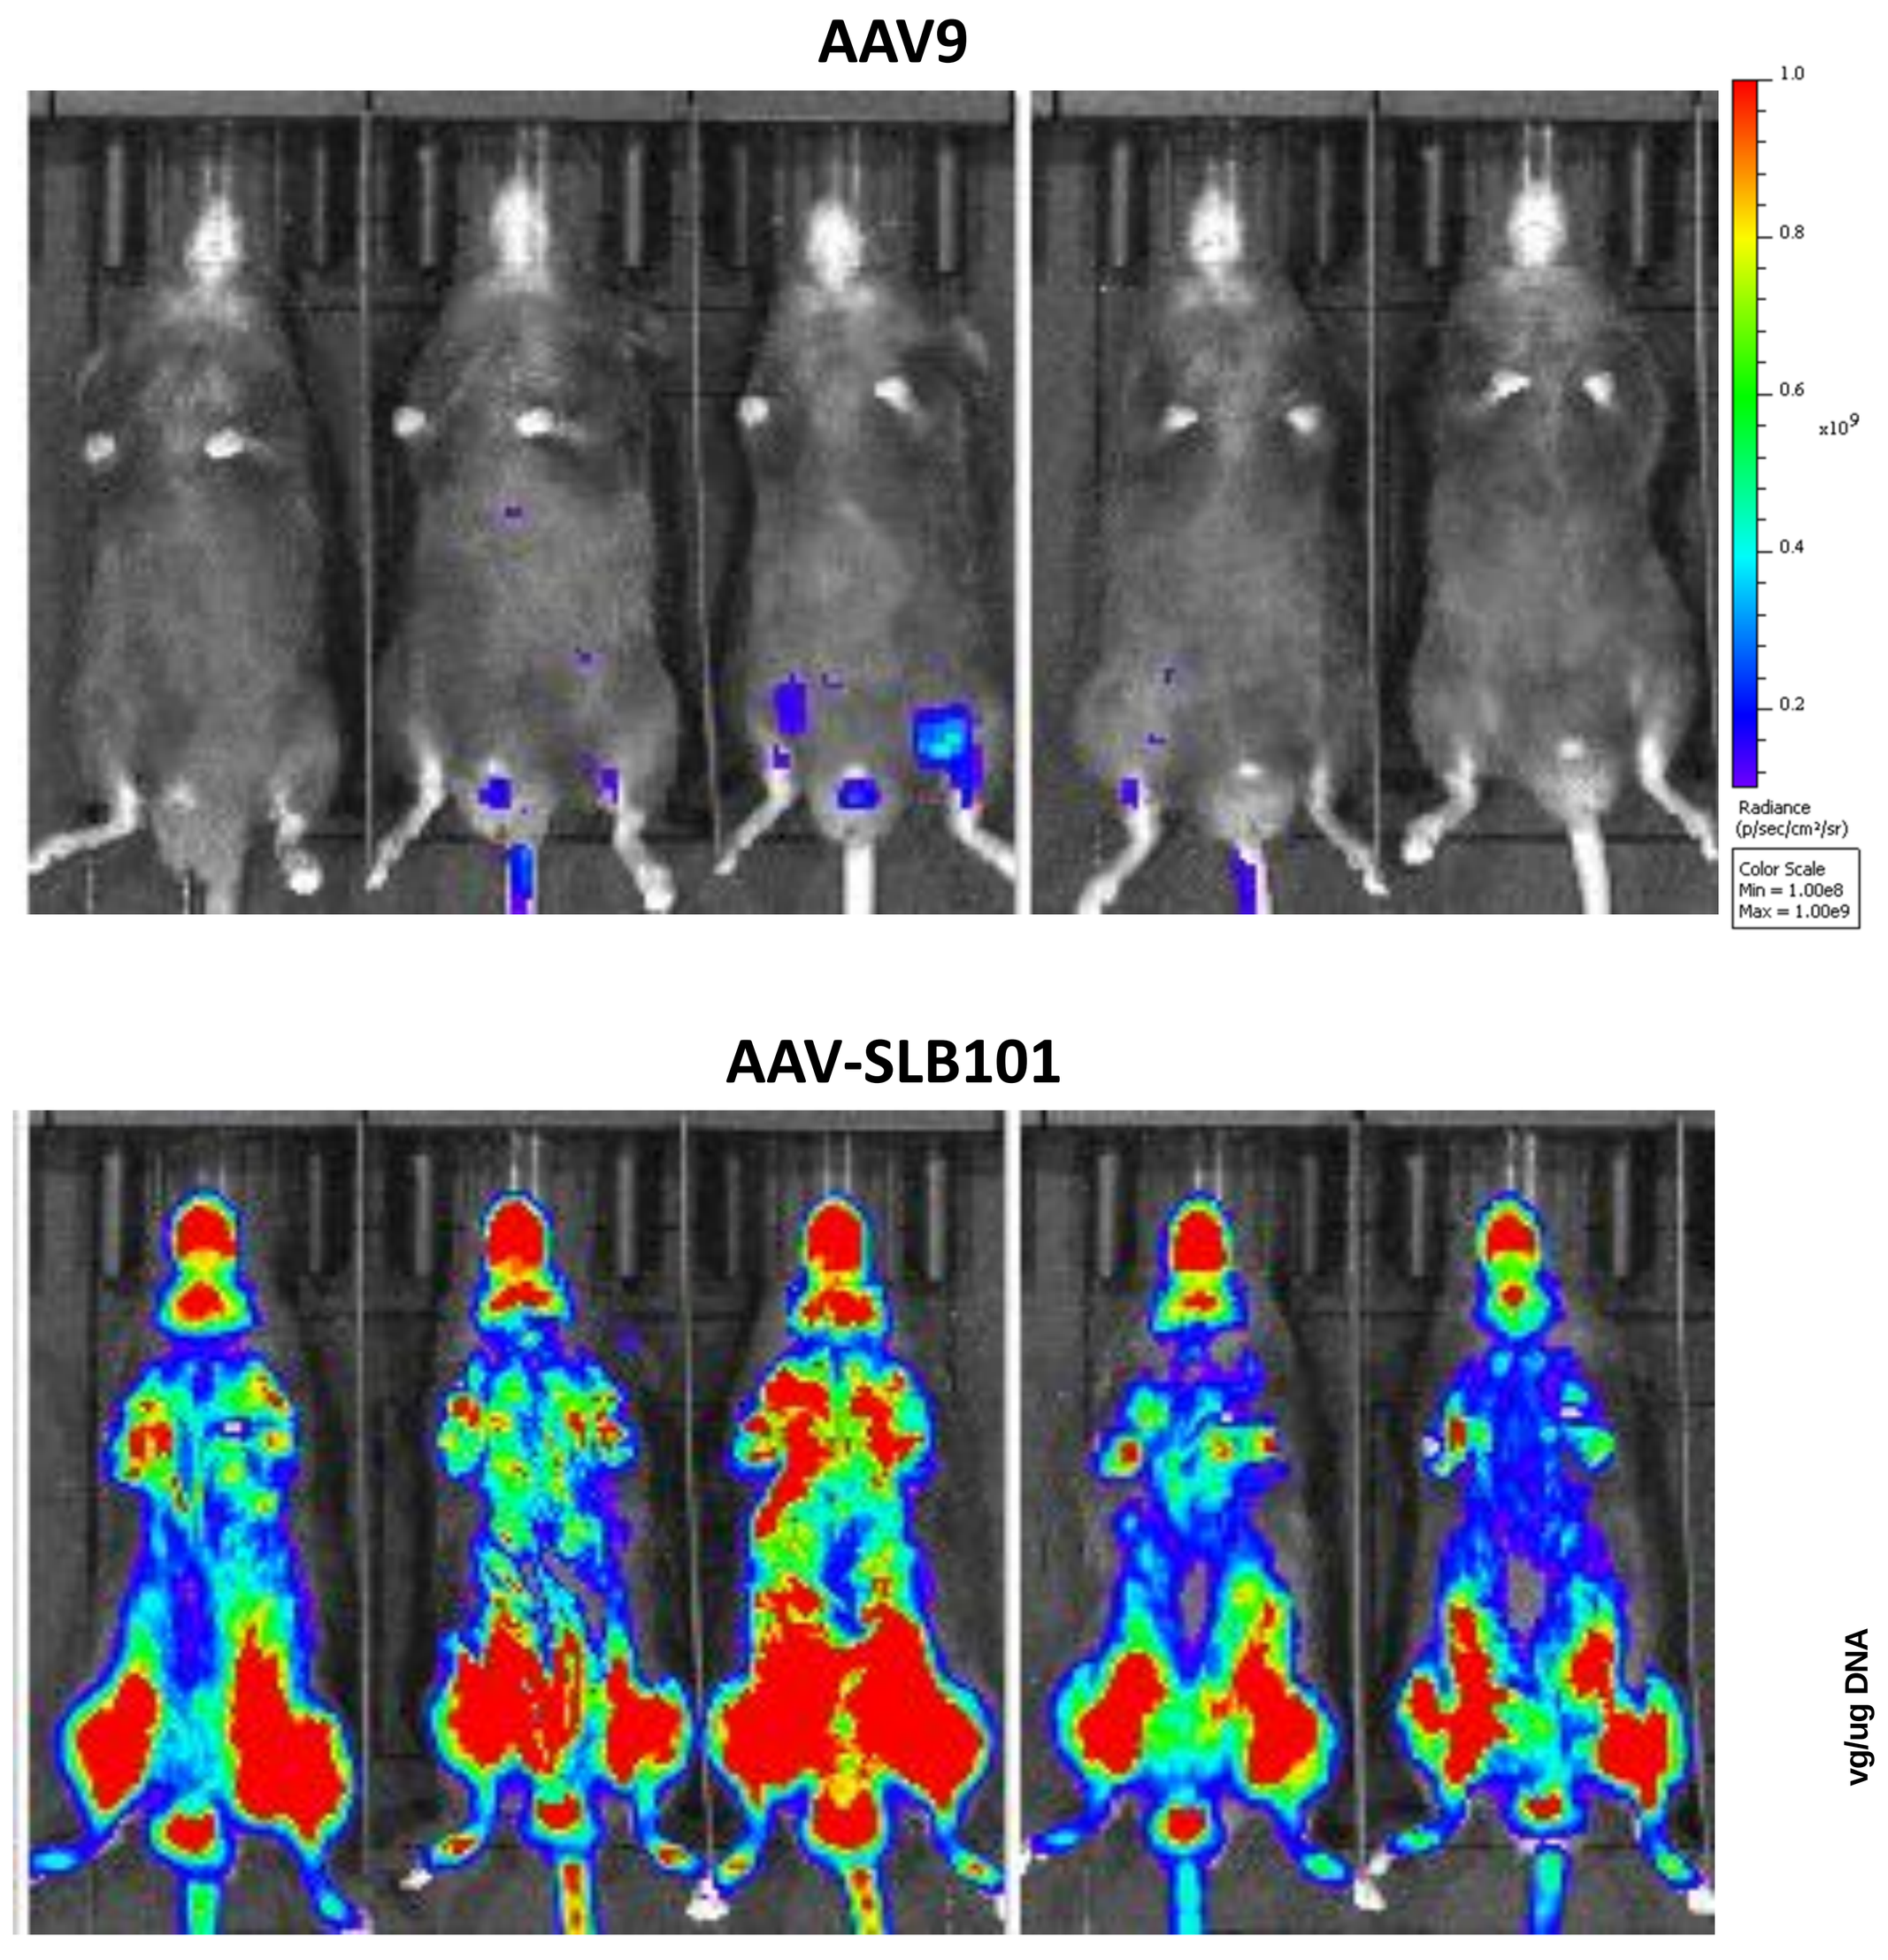
AAV9
AAV-SLB101

Figure 2. C57BL/6 mice, 5-6 weeks old, were systemically injected with AAV9 and AAV-SLB101 at a dose of 5E13 vg/kg. Mice were imaged weekly for luciferase expression using an in vivo imaging system (IVIS) then necropsied four weeks post-injection for quantification of vector biodistribution and luciferase expression in individual tissues. Representative whole body IVIS images at four weeks post-injection are shown on the left. Biodistribution of AAV9 vs. AAV-SLB101 in quadriceps and liver are shown in graphs below, with statistics determined by Welch's t test ($p < 0.05$). Data is consistent with biodistribution observed in *DMD[mdx]* mice in both tissues.

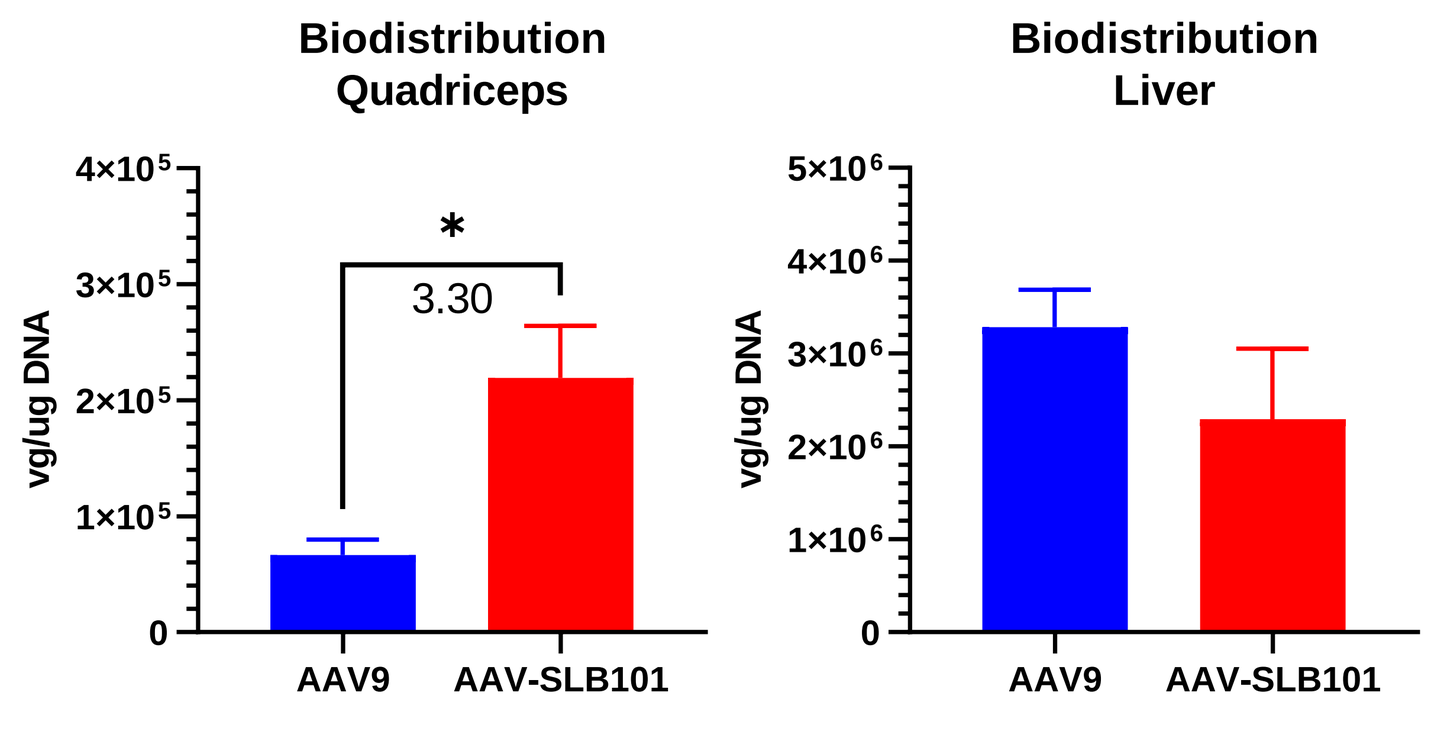
Biodistribution Quadriceps
Biodistribution Liver
vg/ug DNA
3.30
AAV9
AAV-SLB101

Novel Capsid in NHPs

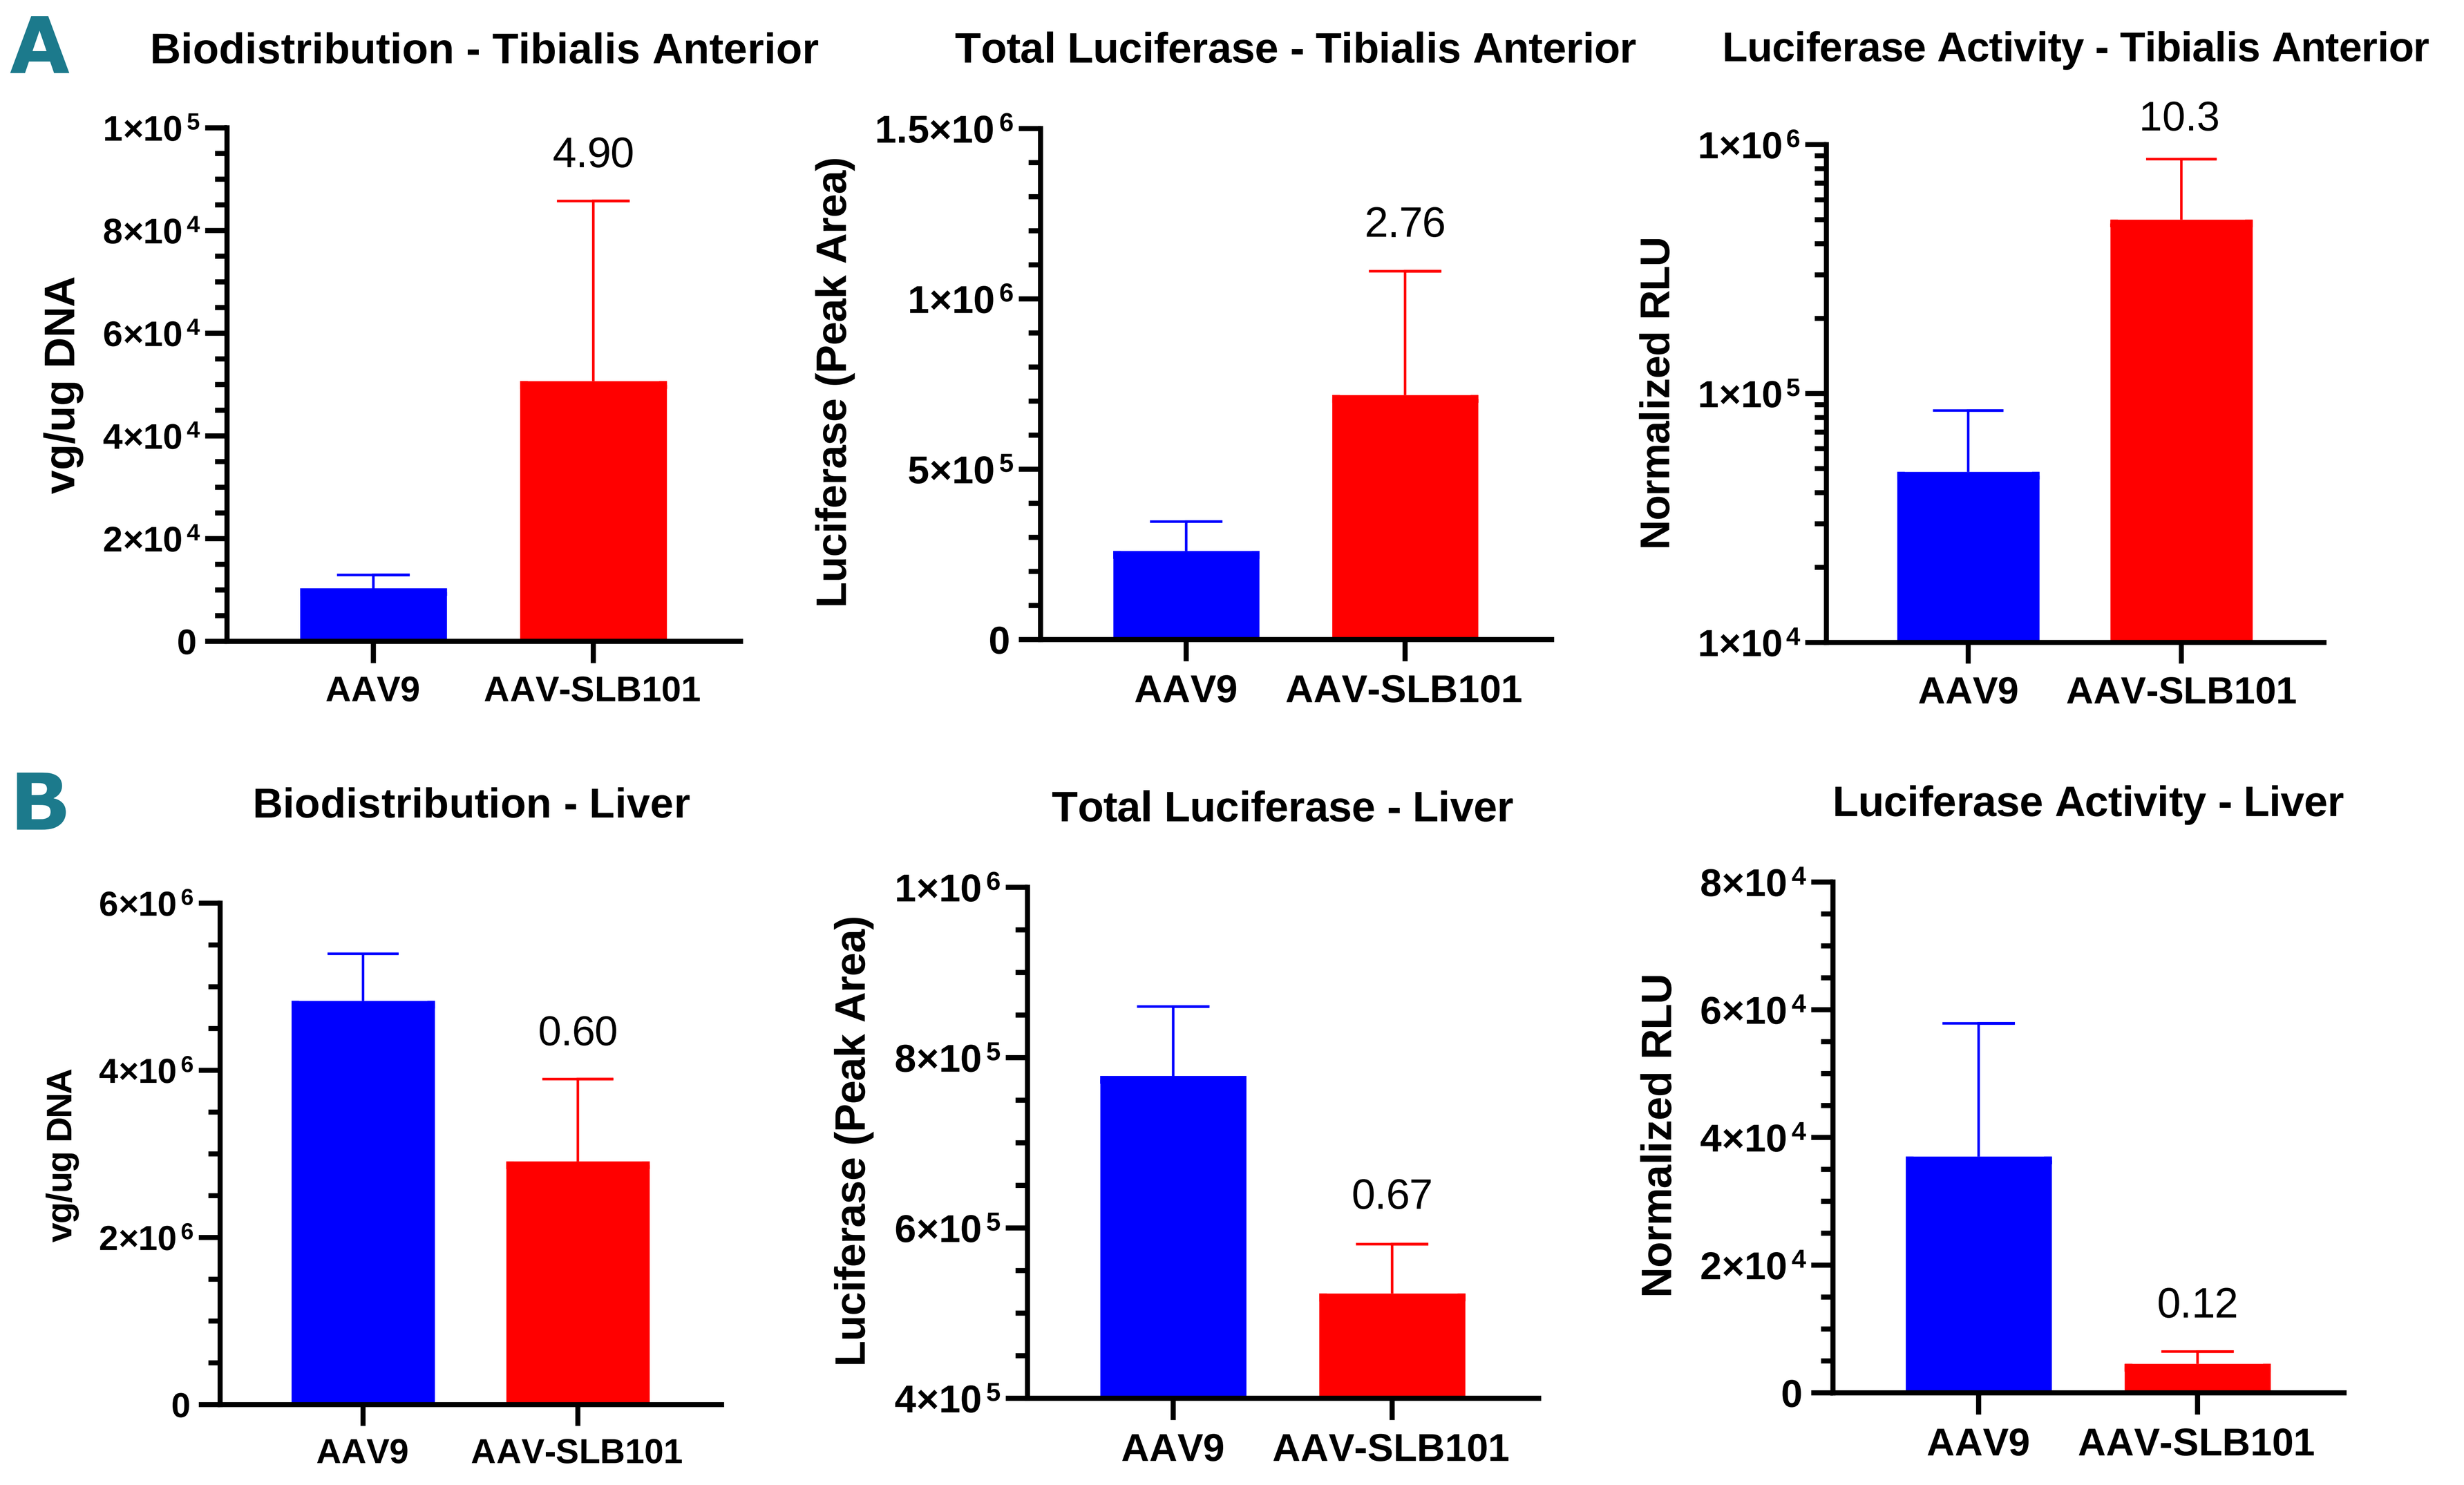
A
Biodistribution - Tibialis Anterior
Total Luciferase - Tibialis Anterior
Luciferase Activity - Tibialis Anterior
4.90
2.76
10.3
B
Biodistribution - Liver
Total Luciferase - Liver
Luciferase Activity - Liver
0.60
0.67
0.12
vg/ug DNA
Luciferase (Peak Area)
Normalized RLU
AAV9
AAV-SLB101

Figure 3. Juvenile cynomolgus macaques, approximately 2 years old, were systemically infused with AAV9 and AAV-SLB101 at 5E13 vg/kg. Two males and two females were dosed in each group. Animals were necropsied four weeks post-injection and tissues were harvested for biodistribution, total luciferase protein and luciferase activity. Fold change versus AAV9 is indicated on each graph.

A. Increased biodistribution, total luciferase expression and luciferase activity of AAV-SLB101 in comparison to AAV9 in skeletal muscle were observed.
B. Decreased biodistribution, total luciferase expression and luciferase activity of AAV-SLB101 in comparison to AAV9 in liver were observed.

Mechanism of Action

Confirmation of hypothesized binding partner

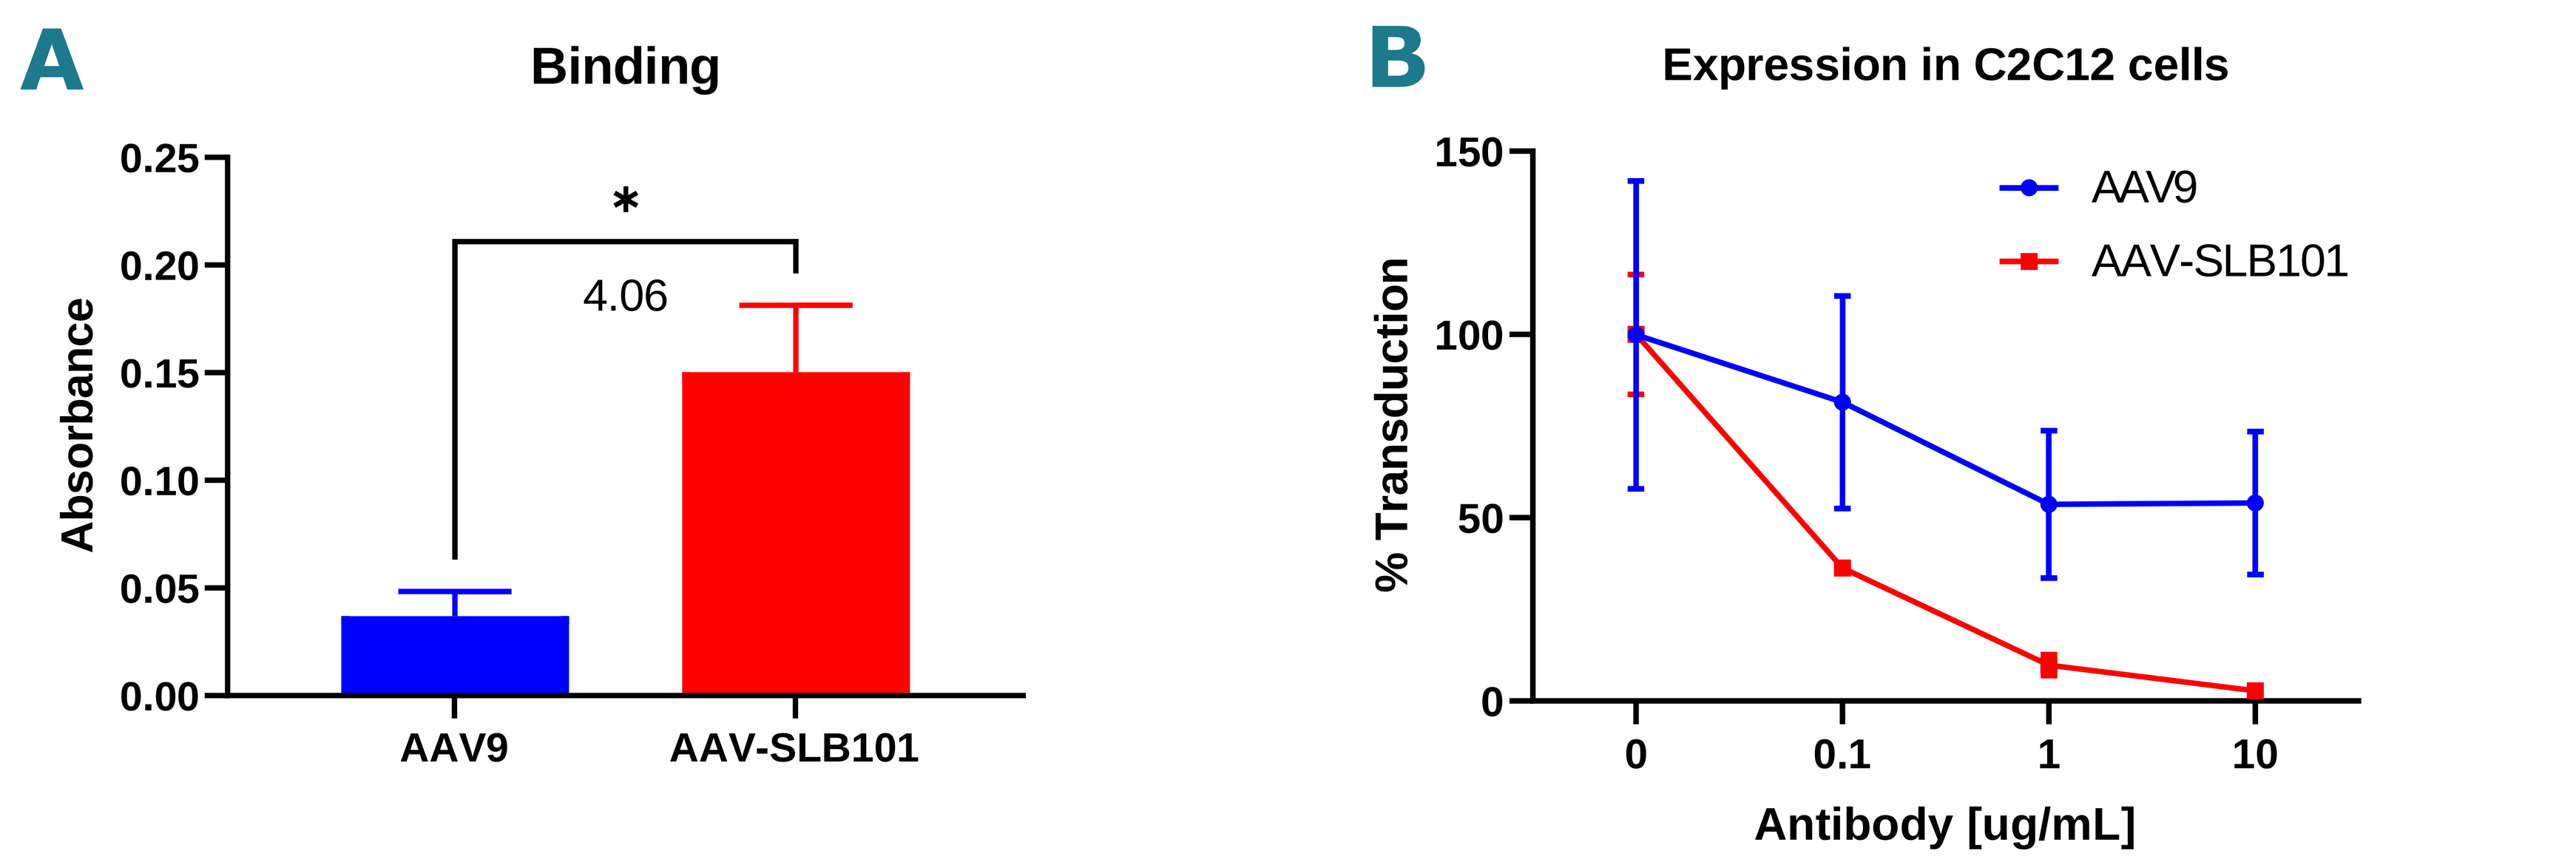
A
Binding
Absorbance
4.06
AAV9
AAV-SLB101
B
Expression in C2C12 cells
% Transduction
Antibody [ug/mL]
AAV9
AAV-SLB101

Figure 4. A binding partner of AAV-SLB101 was identified by bioinformatics to model binding in silico and further explored *in vitro*.

A. Assays were developed to measure binding of AAV9 and AAV-SLB101 to a potential binding partner of AAV-SLB101. Binding of AAV-SLB101 was measured to be 4x higher than AAV9 to this protein. Statistics are determined by Welch's t test (* $p < 0.05$).
B. Transduction of C2C12 cells with AAV-SLB101 was inhibited by pre-incubation with an antibody against the AAV-SLB101 binding partner in comparison to AAV9.

Rational Design Based on MoA

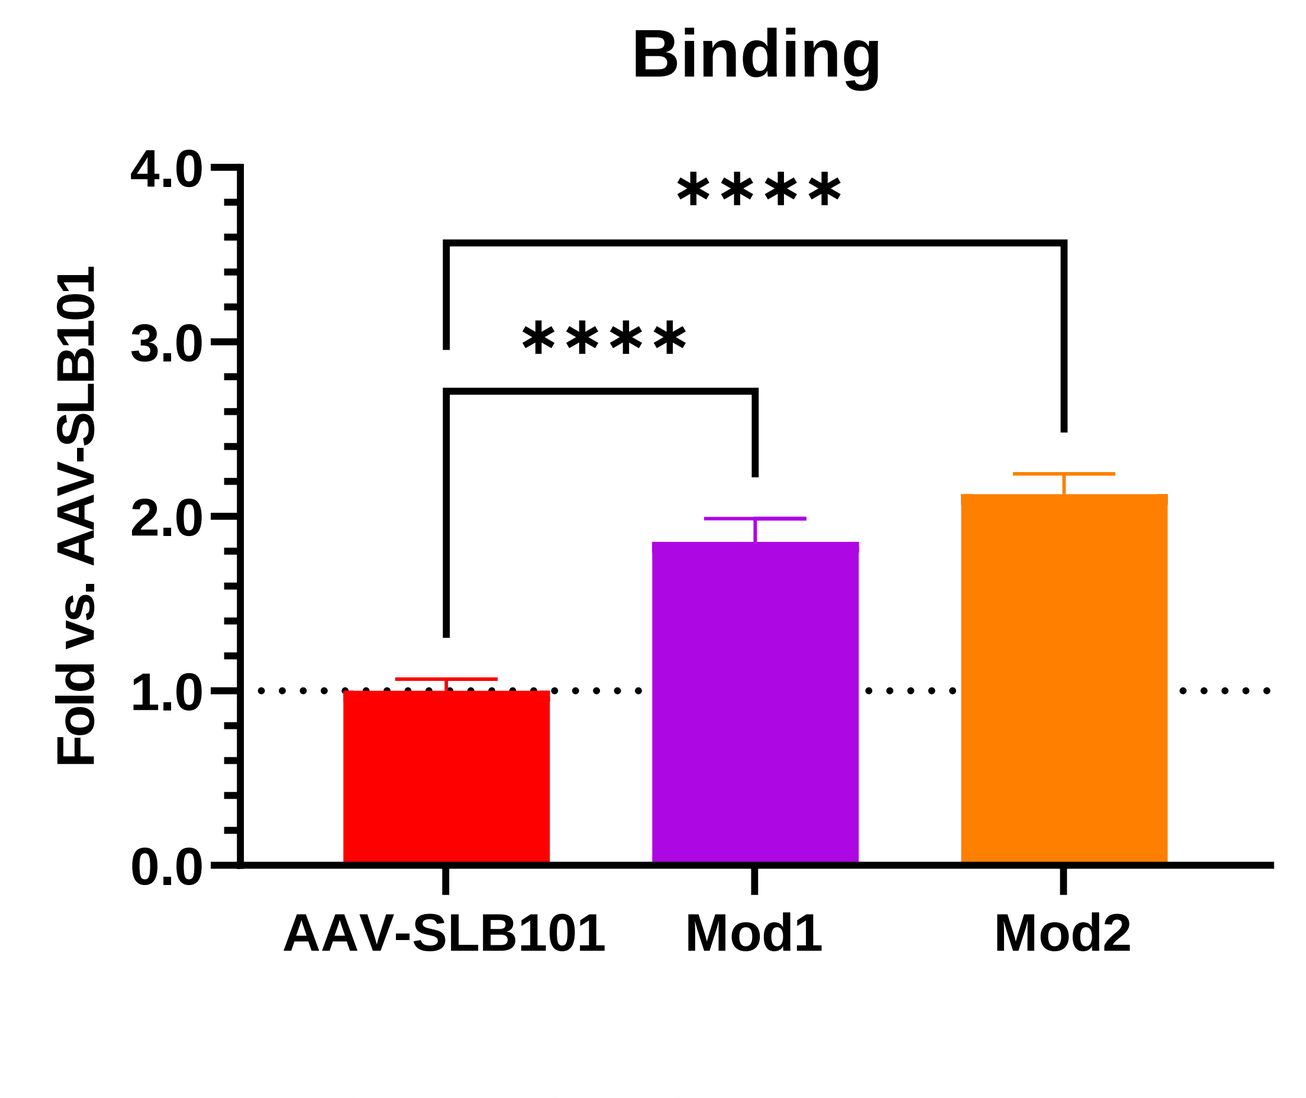
Binding
Fold vs. AAV-SLB101
AAV-SLB101
Mod1
Mod2

Figure 5. Based on other known interactions with this protein, additional novel capsids were rationally designed to further increase binding. Mod1 and Mod2 both had statistically significant increases in binding to this protein over AAV-SLB101 (**** $p < 0.0001$) as determined by ordinary one-way ANOVA.

Potential implications

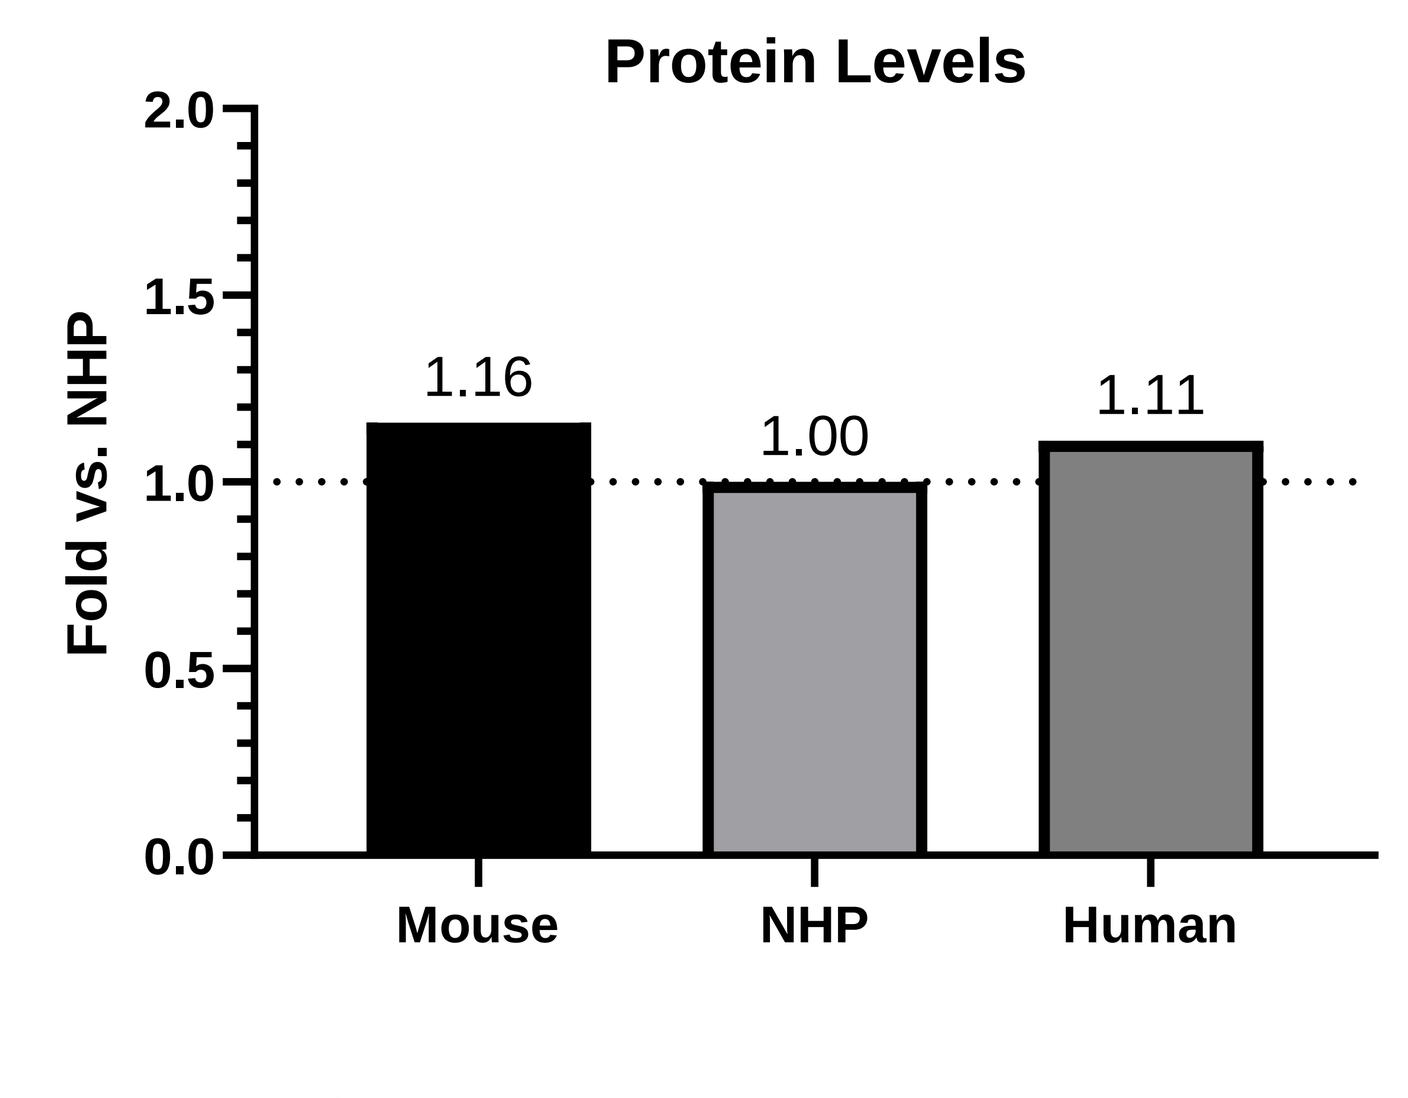
Protein Levels
Fold vs. NHP
1.16
1.00
1.11
Mouse
NHP
Human

Figure 6. This protein was detected in mouse, non-human primate and human muscle in similar abundance. Fold change relative to NHPs is indicated above each bar on the graph.

Conclusions

- A novel capsid, AAV-SLB101, has continued to show superior transduction efficiency in comparison to AAV9 in both wild type and *DMD[mdx]* mice, as well as non-human primates.
- Increased biodistribution to skeletal muscle and decreased biodistribution to liver of AAV-SLB101 in comparison to AAV9 remains consistent across animal models tested and correlates to changes in protein expression of both microdystrophin (increased in muscle) and a luciferase reporter (increased in muscle and decreased in liver).
- MoA studies identified a binding partner of AAV-SLB101, providing a basis for additional capsid modifications to potentially increase binding across multiple species.
- Solid's SGT-003 program incorporates additive improvements from both the use of the novel muscle-tropic capsid AAV-SLB101 and triple transfection manufacturing to result in multiple-fold higher levels of microdystrophin expression as compared to the first-generation candidate SGT-001 produced by HSV-based manufacturing.