Novel AAV Capsid Identification and Characterization for Neuromuscular and Cardiac Indications

Jennifer Green, Jessica F Boehler, Meghan Soustek-Kramer, Jamie L Marshall, Tiffany Willacy, Prushti Bhavsar, Kristy J Brown, Sharon McGonigle, Mariah Prom*, Margaret Beatka*, Emily Ott*, Michael W Lawlor*, Carl Morris

*Solid Biosciences, Charlestown, MA, USA; * Diverge Translational Science Laboratory, Milwaukee, WI, USA*

Introduction

- Adeno-associated virus (AAV) mediated gene therapy continues to be a promising therapeutic path for various diseases. However, high systemic doses currently required to achieve widespread therapeutic benefit in cases like Duchenne muscular dystrophy (DMD) can pose potential safety risks, which could be decreased or eliminated if therapeutic benefit could be achieved using lower doses through a more targeted and efficacious vector.
- Previously, we described a lead novel AAV capsid of interest (AAV-SLB101) that has increased muscle tropism and decreased liver transduction in both disease and non-disease contexts in multiple animal models (*DMD^{mdx}* mice, wild type mice and non-human primates).
- We are continuing to use rational design, structure activity relationship (SAR)-like approaches to iterate and test additional novel AAV capsids with:
 1. *Modifications based on the mechanism of action of AAV-SLB101*
 2. *Combinations of AAV-SLB101 and other modifications to create a synergistic effect*
 3. *Modifications based on muscle biology*

Mechanism of Action – Confirmation of Binding Partner

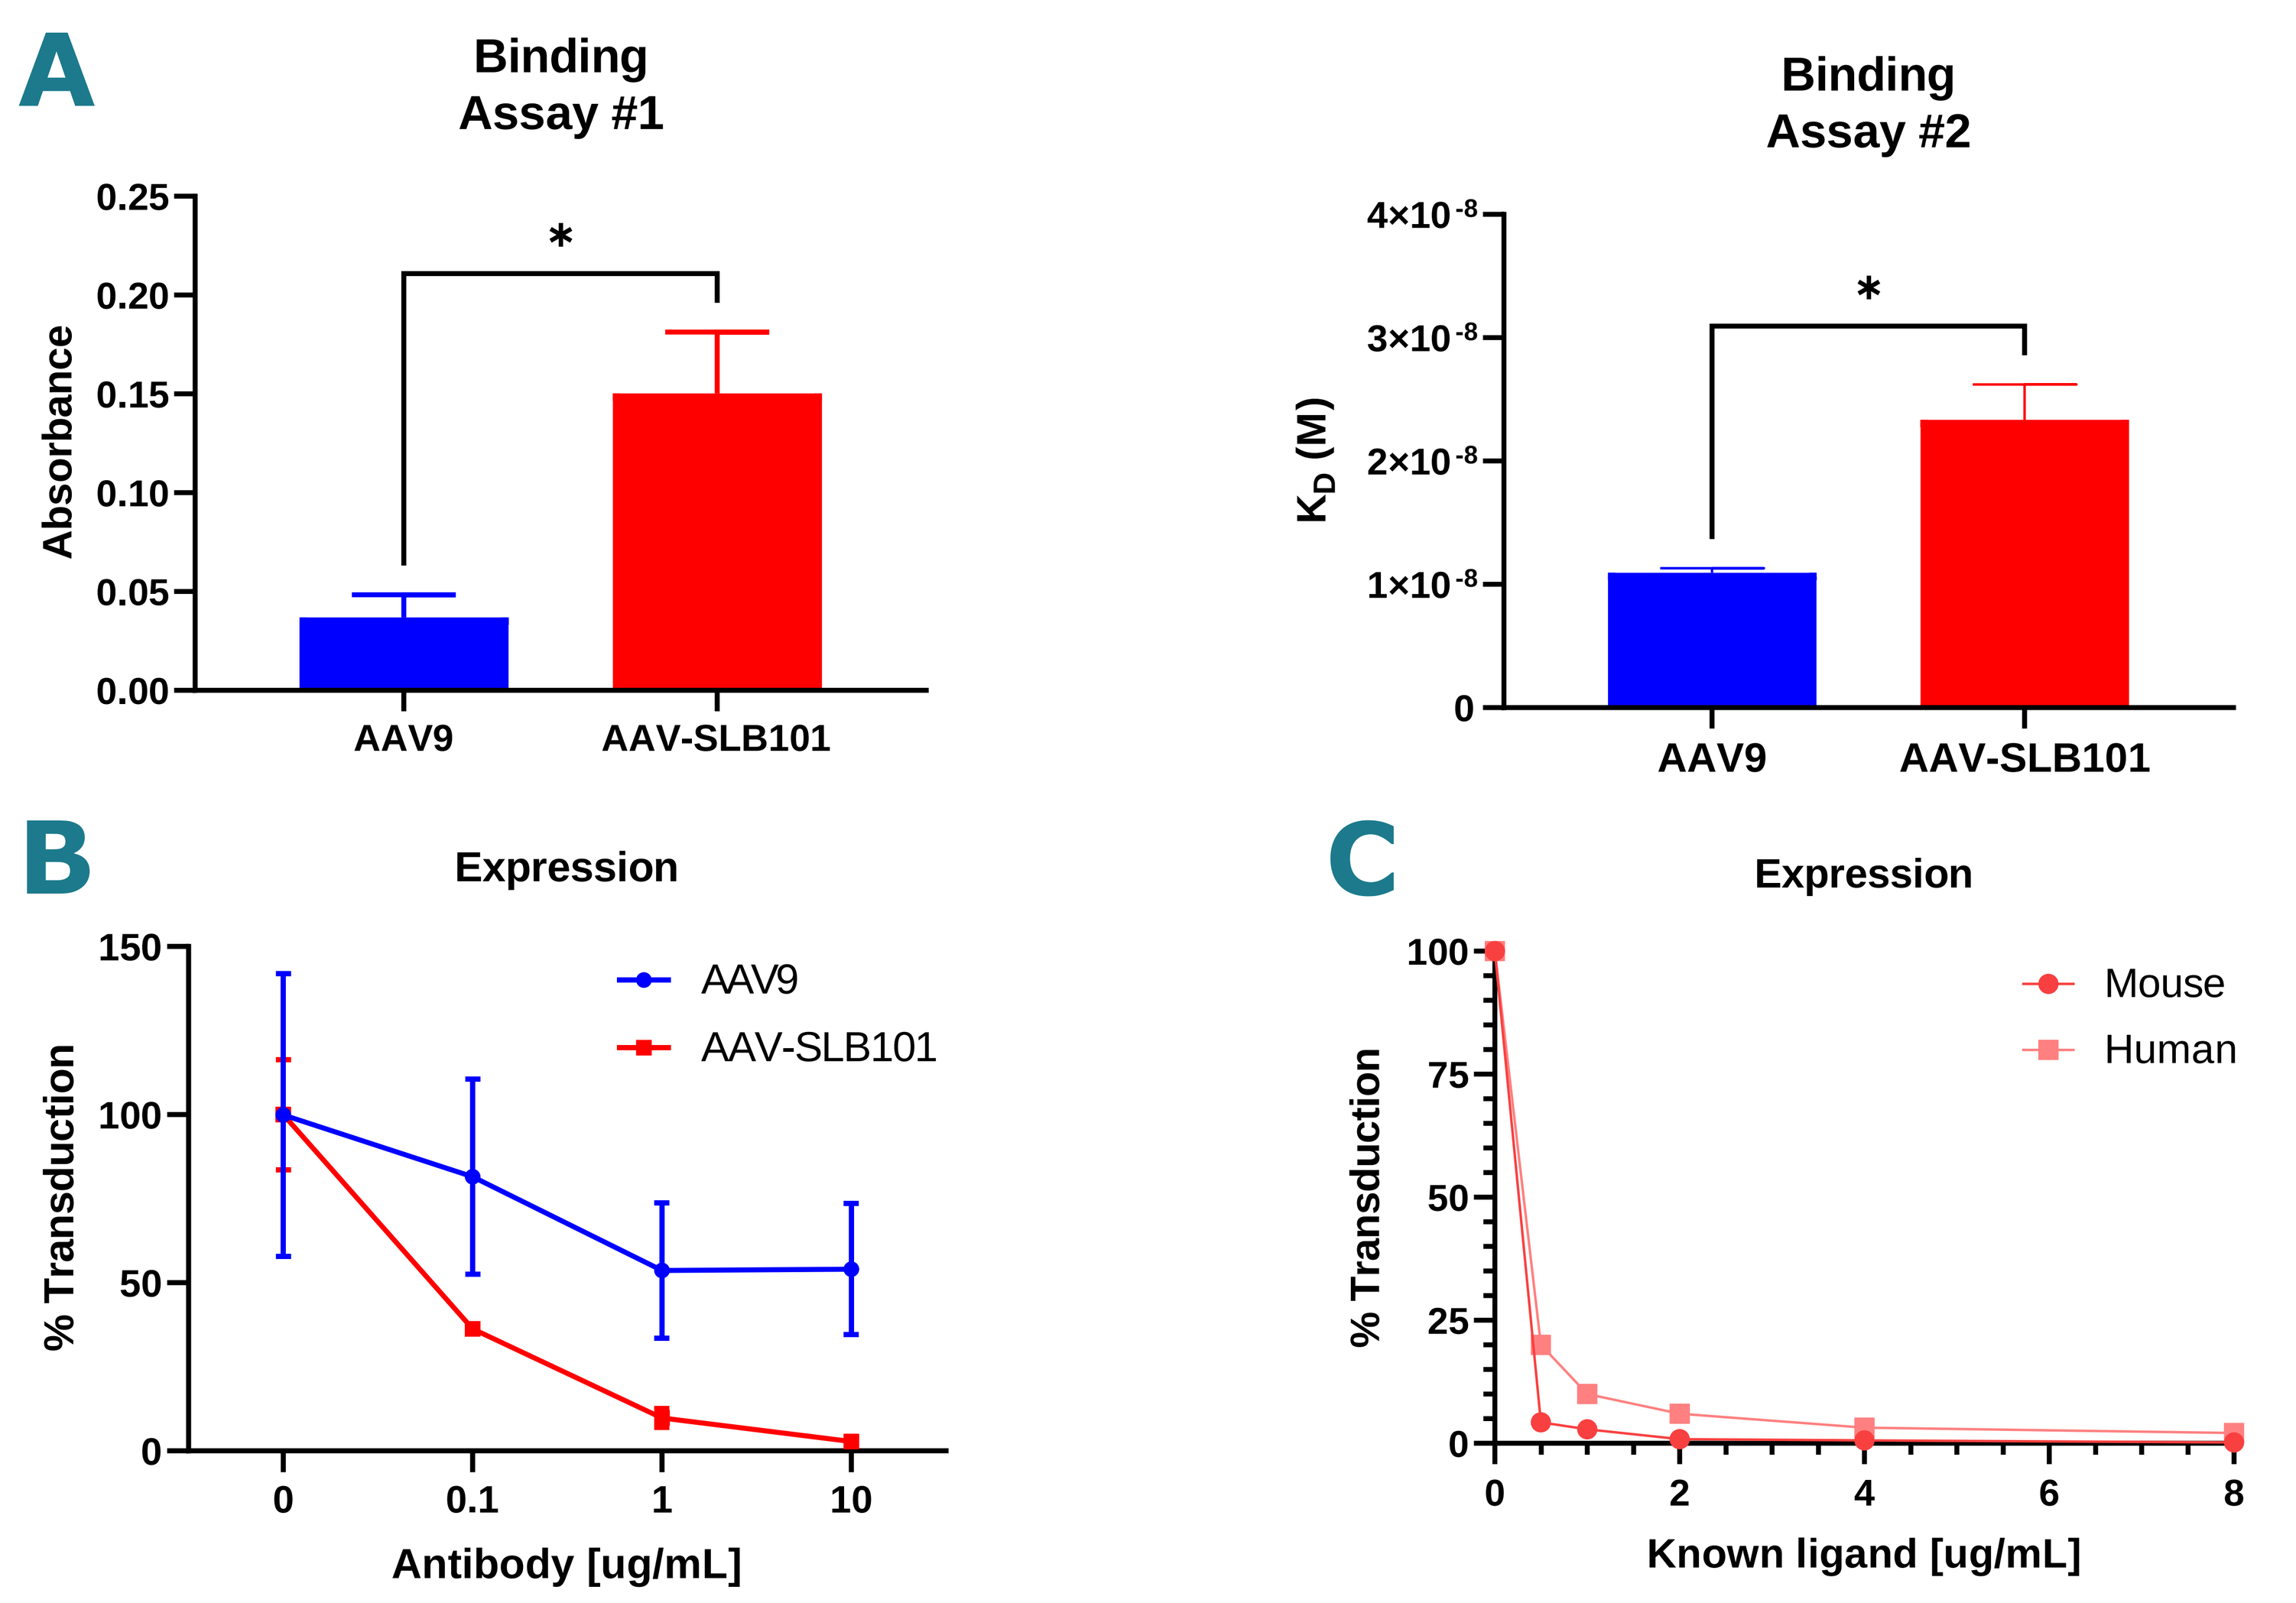
A
Binding Assay #1
Absorbance
AAV9
AAV-SLB101
Binding Assay #2
K_D (M)
B
Expression
% Transduction
Antibody [ug/mL]
C
Expression
Mouse
Human
Known ligand [ug/mL]

Figure 1. A binding partner of AAV-SLB101 was identified by bioinformatics modeling binding in silico and further explored *in vitro*. This binding partner is expressed at similar levels in mice, non-human primates and humans (data not shown), increasing the potential of AAV-SLB101 translation from nonclinical models to a humans.

A. Two independent assays were developed to measure binding of AAV to recombinant versions of the predicted binding partner of AAV-SLB101. Binding of AAV-SLB101 was measured to be higher than AAV9 to this protein in both assays. Statistics were determined by Welch's t test (* $p < 0.05$).
B. Transduction of mouse skeletal muscle cells with AAV-SLB101 was inhibited by pre-incubation with an antibody against the AAV-SLB101 binding partner in comparison to AAV9.
C. Transduction of both mouse and human skeletal muscle cells with AAV-SLB101 was also inhibited by pre-incubation of the cells with a known ligand to AAV-SLB101's binding partner.

MoA - Characterization of Mod1 and Mod2 *In Vitro* & *In Vivo*

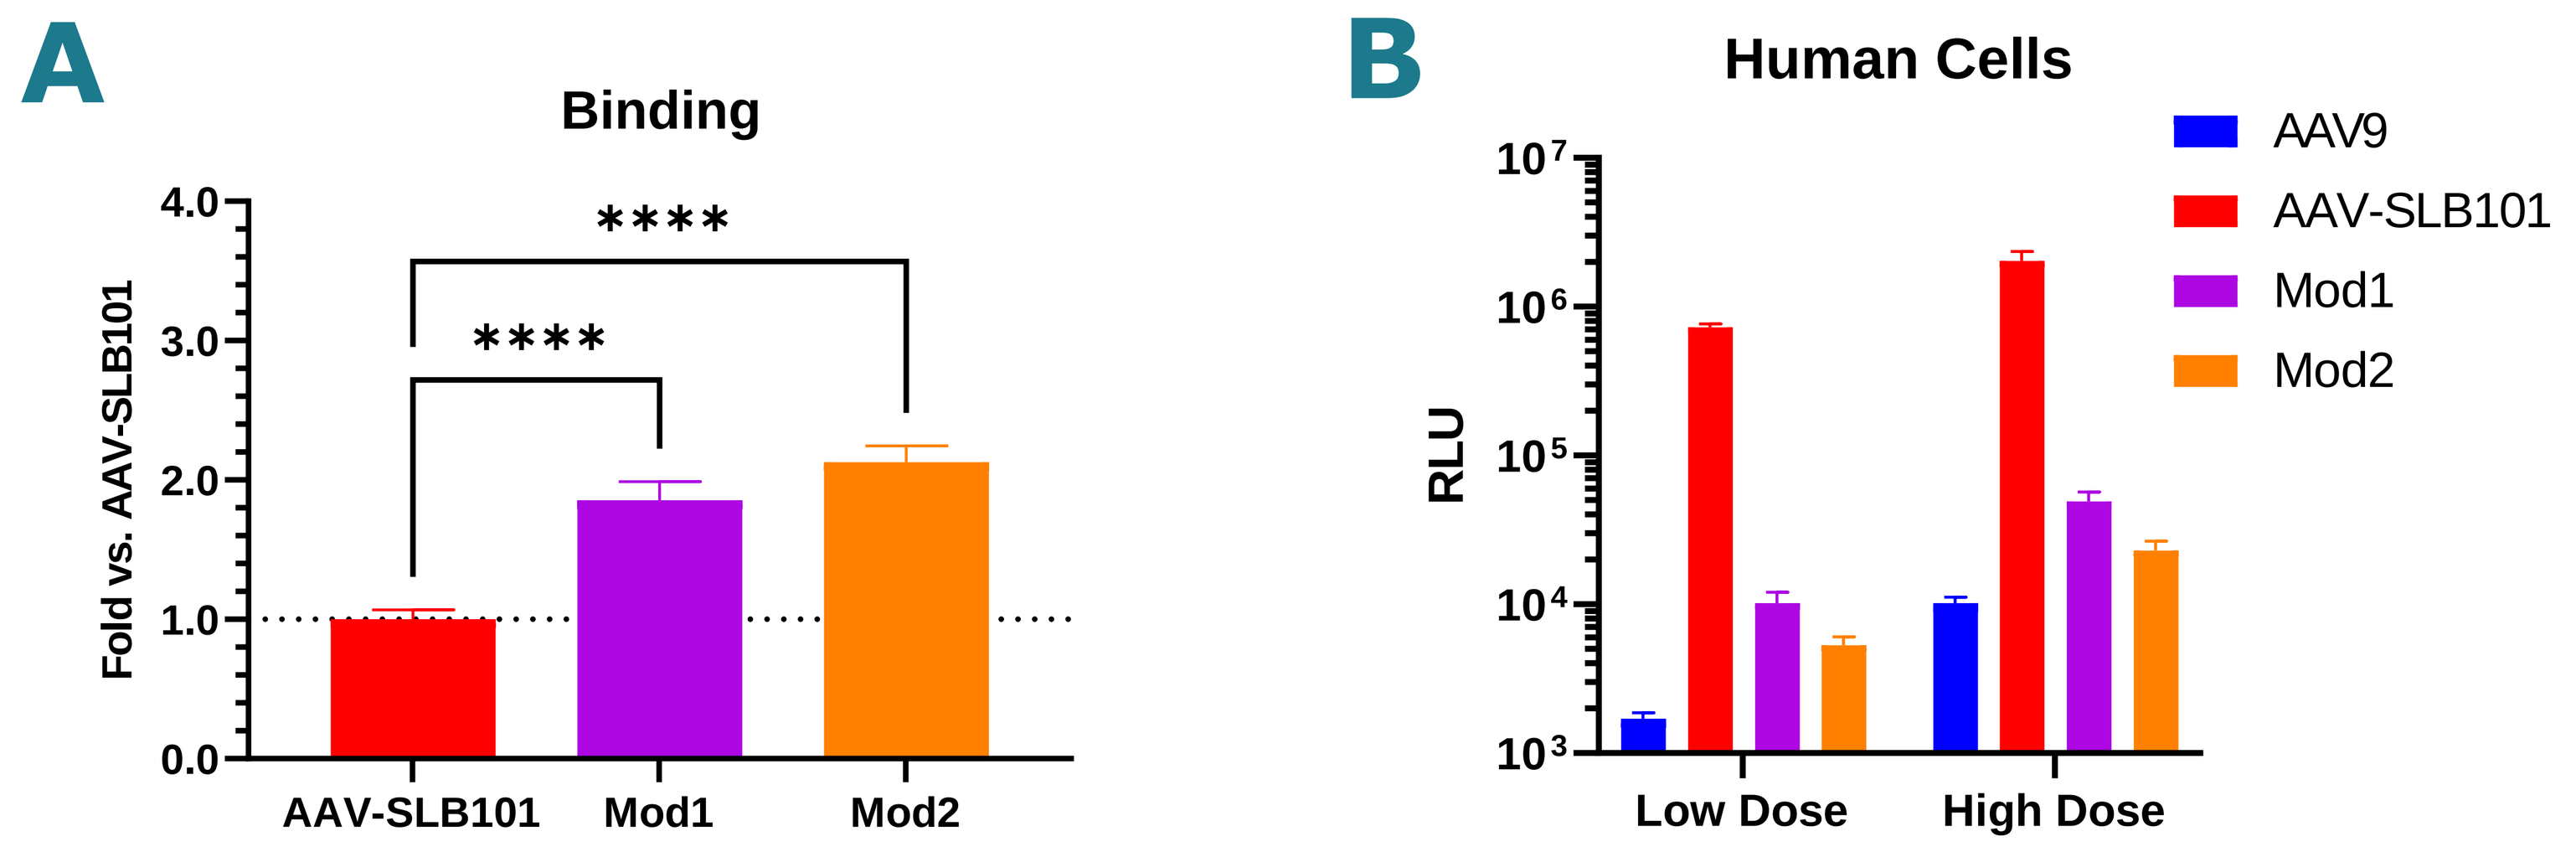
A
Binding
Fold vs. AAV-SLB101
AAV-SLB101
Mod1
Mod2
B
Human Cells
RLU
AAV9
AAV-SLB101
Mod1
Mod2
Low Dose
High Dose

Figure 2. Based on a known ligand of the identified binding partner, additional novel capsids were rationally designed to further increase binding.

A. Mod1 and Mod2 both had statistically significant increases in binding to the human recombinant protein over AAV-SLB101, as determined by ordinary one-way ANOVA (**** $p < 0.0001$).
B. However, this increased binding did not lead to increases in transduction, as measured by luciferase activity 48 hours after transduction of human DMD skeletal muscle cells.

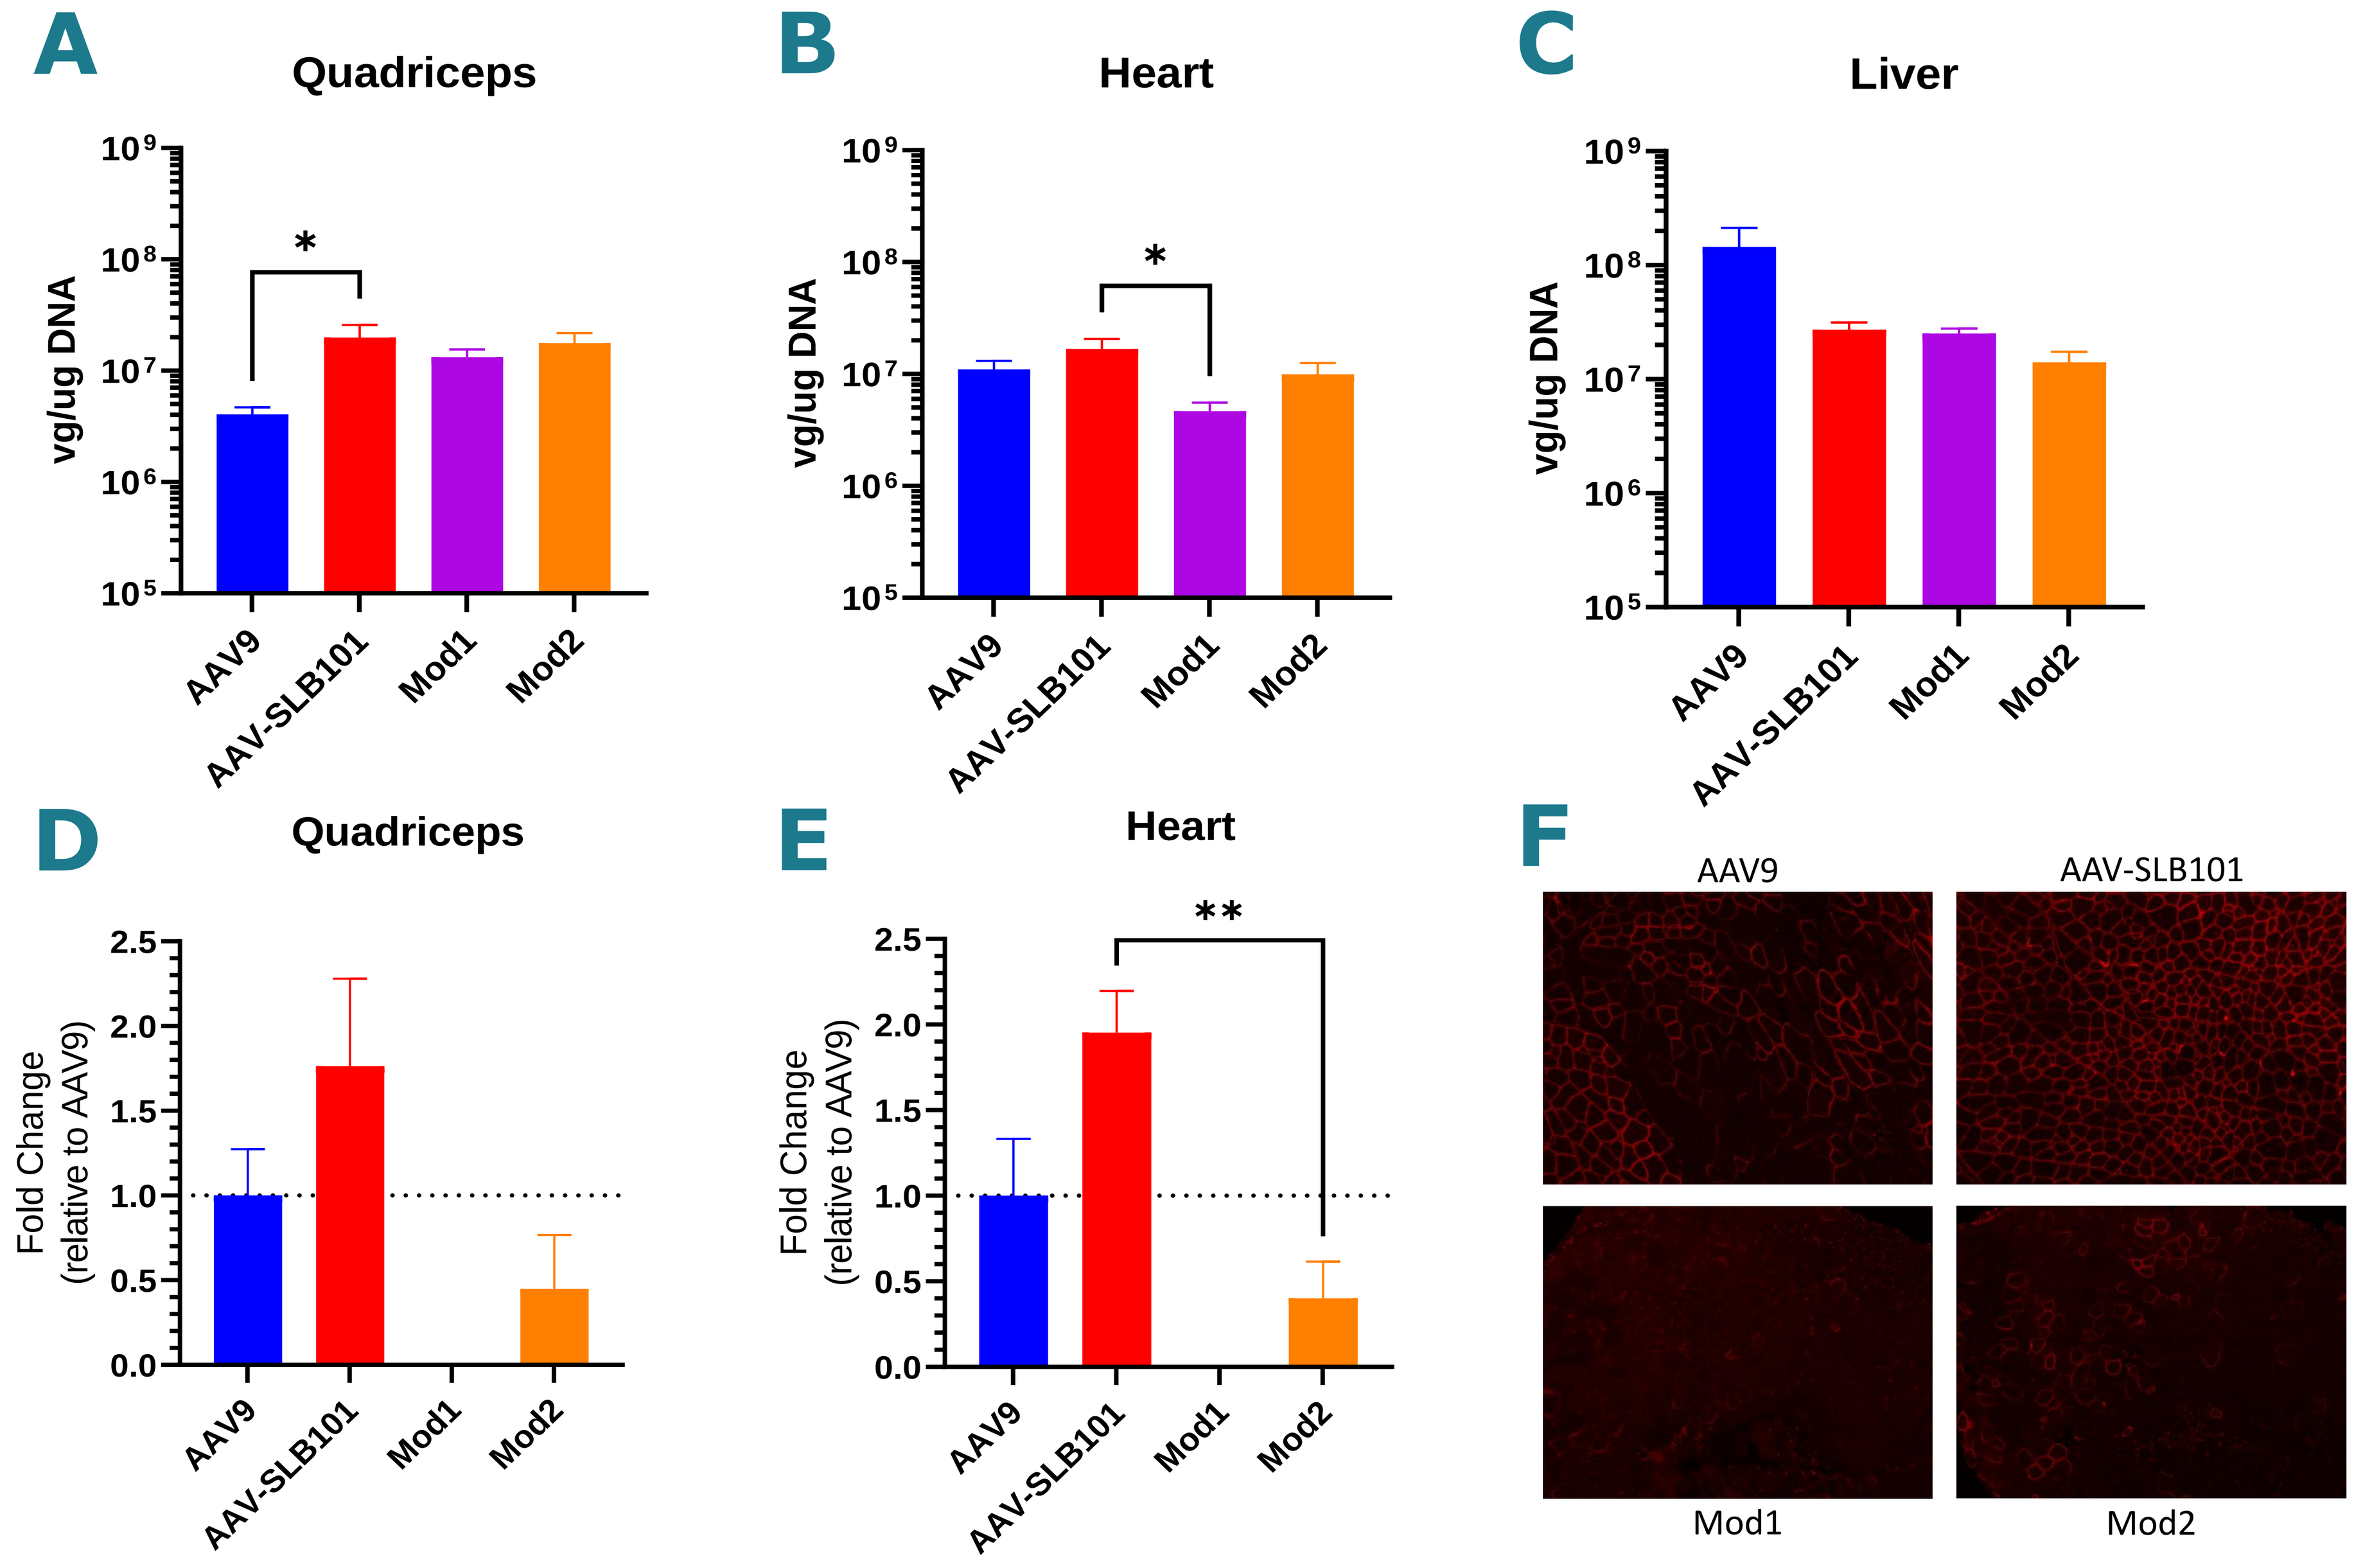
A
Quadriceps
B
Heart
C
Liver
vg/ug DNA
D
Quadriceps
E
Heart
Fold Change (relative to AAV9)
AAV9
AAV-SLB101
Mod1
Mod2
F
AAV9
AAV-SLB101
Mod1
Mod2

Figure 3. Mod1 and Mod2 resulted in A) comparable biodistribution to mouse skeletal muscle in comparison to AAV-SLB101 and B) similar or statistically significantly lower biodistribution to mouse cardiac muscle in comparison to AAV-SLB101 in *DMD^{mdx}* mice two weeks after a 5E13 vg/kg dose was delivered IV via tail vein. AAV-SLB101, Mod1 and Mod2 resulted in lower biodistribution to mouse liver (C), but those differences were not statistically significant. Despite similarities in biodistribution, Mod1 and Mod2 resulted in lower microdystrophin expression than AAV-SLB101 in both skeletal (D) and cardiac (E) muscle. Statistics for each graph were determined by ordinary one-way ANOVA (* $p < 0.05$, ** $p < 0.01$). F) Representative immunofluorescent images of quadriceps from each treatment.

Combination Variants - *In Vitro* Transduction

Figure 4. Synergistic variants, Mod3-5, were produced to combine the novel capsid modification from AAV-SLB101 with additional modifications in other locations in the capsid predicted to further de-target the liver or increase tropism to muscle. Mod5 resulted in statistically significant increases in microdystrophin expression over AAV9, similar to AAV-SLB101, as measured 72 hours after transduction of mouse skeletal muscle cells. Statistics were determined by ordinary one-way ANOVA (**** $p < 0.0001$).

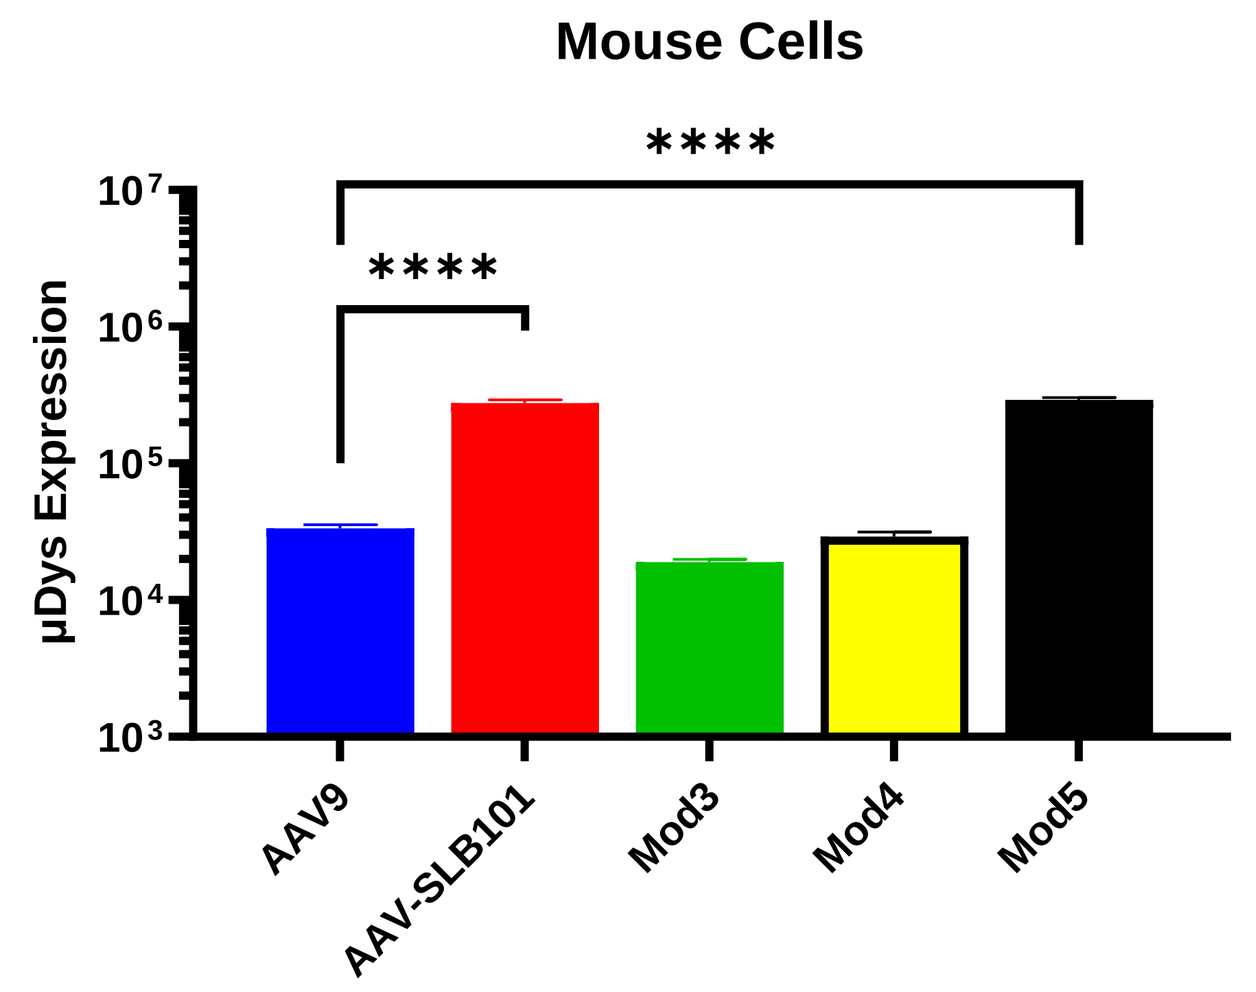
Mouse Cells
µDys Expression
AAV9
AAV-SLB101
Mod3
Mod4
Mod5

Muscle Biology-Based Modification *In Vitro* & *In Vivo*

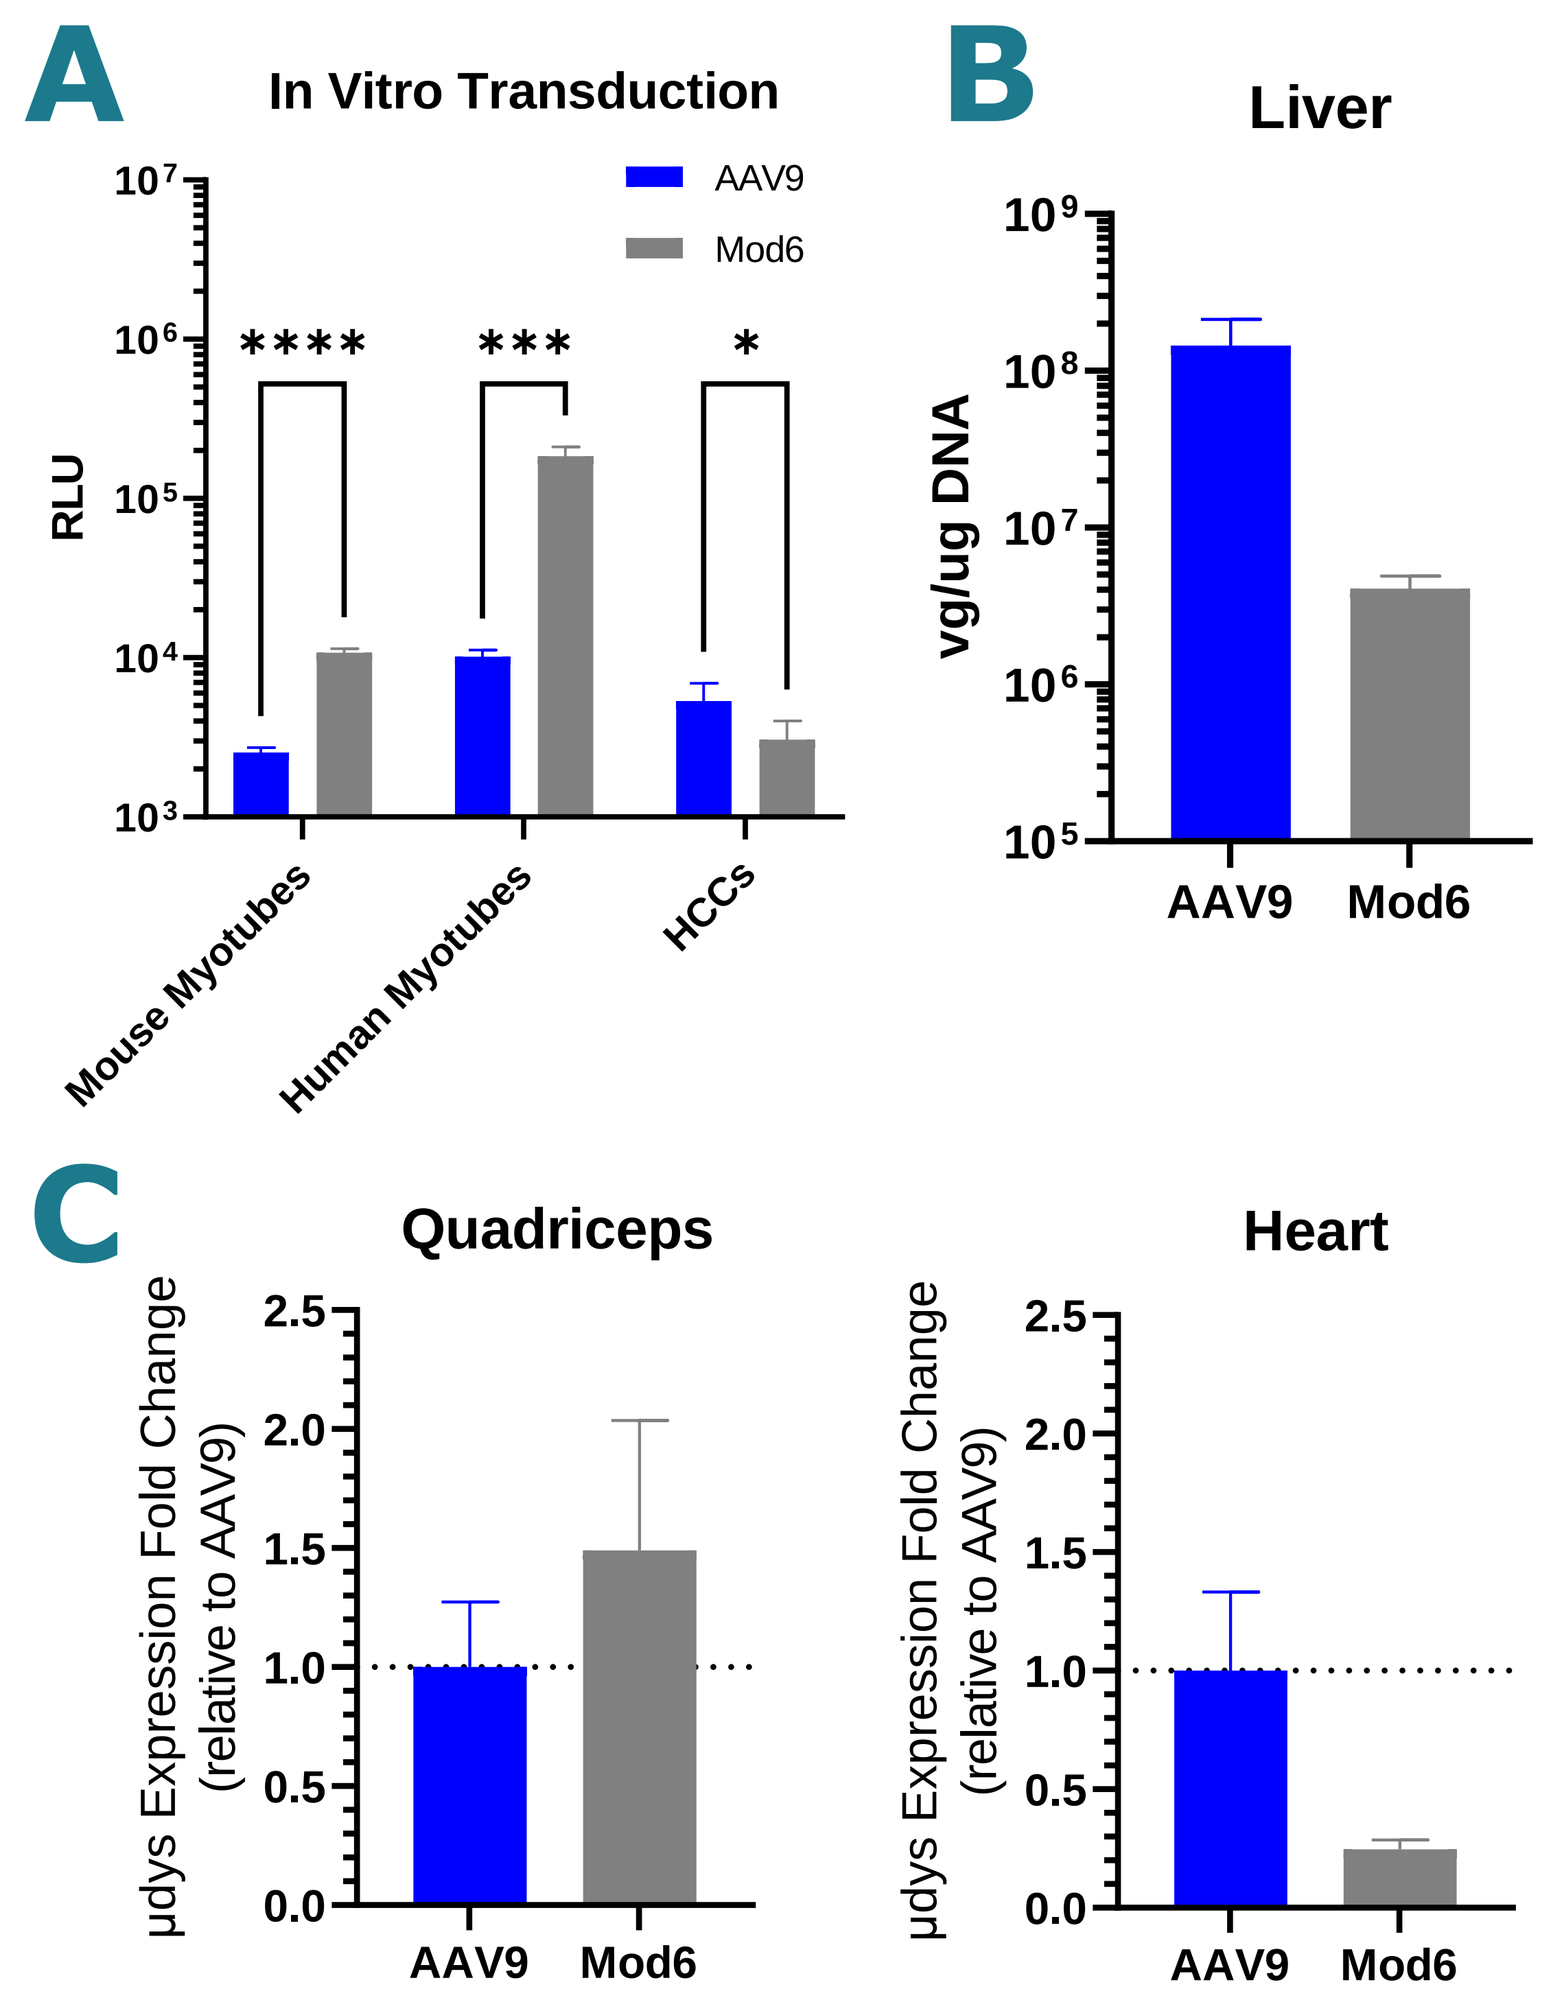
A
In Vitro Transduction
AAV9
Mod6
RLU
Mouse Myotubes
Human Myotubes
HCCs
B
Liver
vg/ug DNA
C
Quadriceps
Heart
µdys Expression Fold Change (relative to AAV9)

Figure 5. Another novel capsid was compared to AAV9 *in vitro* and *in vivo* for skeletal muscle cell transduction and liver cell de-targeting.

A. Mod6 had statistically significant increases in transduction of both mouse (**** $p < 0.0001$) and human (*** $p < 0.001$) skeletal muscle cells and decreases in transduction of human hepatocellular carcinoma cells (* $p < 0.05$) in comparison to AAV9, as measured by luciferase activity 48-72 hours after transduction. Statistics were determined by multiple unpaired t tests.
B. In *DMD^{mdx}* mice two weeks after a 5E13 vg/kg dose was delivered IV via tail vein, Mod6 resulted in ~35x decreased biodistribution to liver in comparison to AAV9.
C. In the same *DMD^{mdx}* mouse study, Mod6 also resulted in similar microdystrophin expression in quadriceps and trending decreases ($p = 0.06$) in heart in comparison to AAV9. Decreases in heart were predicted due to the expression profile (skeletal muscle > cardiac muscle) of the receptor targeted in this novel AAV capsid.

Conclusions

- Solid's SGT-003 program for DMD incorporates muscle-tropic improvements of the novel capsid, AAV-SLB101.
- Based on the mechanism of action of AAV-SLB101, other known modifications and muscle biology, additional novel AAV capsids have been rationally designed and tested:
 1. *We have shown that increased binding to an AAV-SLB101 receptor does not translate to an increase in transduction.*
 2. *Combinations of modifications have the potential to synergistically further improve specific characteristics in vitro.*
 3. *Different targets can further drive tissue specificity based on predicted receptor expression profiles.*
- Iterations of these SAR-like efforts are continuing to expand our knowledge and understanding of the underlying AAV biology necessary to drive translation from *in vitro* to *in vivo* and tissue-specific tropism.